Next generation N-terminal domain androgen receptor inhibitors with improved potency and metabolic stability in castration-resistant prostate cancer models

Ronan Le Moigne[1], Han-Jie Zhou[1], Nasrin R. Mawji[2], C. Adriana Banuelos[2], Jun Wang[2], Kunzhong Jian[3], Peter Virsik[1], Raymond J. Andersen[3], Marianne D. Sadar[2]

[1]ESSA Pharmaceuticals Inc., Houston, TX, and South San Francisco, USA, [2]Department of Genome Sciences Centre, BC Cancer Agency, 675 West 10th Avenue, Vancouver, BC V5Z 1L3, Canada, [3]Department of Chemistry, University of British Columbia, 2036 Main Mall, Vancouver, BC V6T 1Z1, Canada

Abstract Number: 220

Contact info:
rlemoigne@essapharma.com

BACKGROUND

The androgen receptor (AR) pathway continues to drive most castration-resistant prostate cancer (CRPC) even in late stages of the disease through resistance mechanisms including gain-of-function mutations in the C-terminal ligand-binding domain (LBD) and expression of constitutively active truncated AR splice variants lacking the LBD such as AR-V7. Selective inhibition of the N-terminal domain (NTD) of the AR can inhibit its transcriptional activity even in the presence of anti-androgen resistance.

A Phase I clinical trial of the first-generation AR NTD inhibitor, EPI-506, (a triacetate prodrug of EPI-002 - Ralaniten) demonstrated PSA declines in enzalutamide and/or abiraterone resistant metastatic CRPC patients. However, these declines were less than 50% and of short duration (see abstract 257, poster board L14), revealing the need for more potent and metabolically stable NTD inhibitors.

A new generation of NTD transcriptional inhibitors (Anitens) has been generated. Examples of this new class, demonstrating improved potency, metabolic stability and pharmaceutical properties, will be discussed in this poster.

A

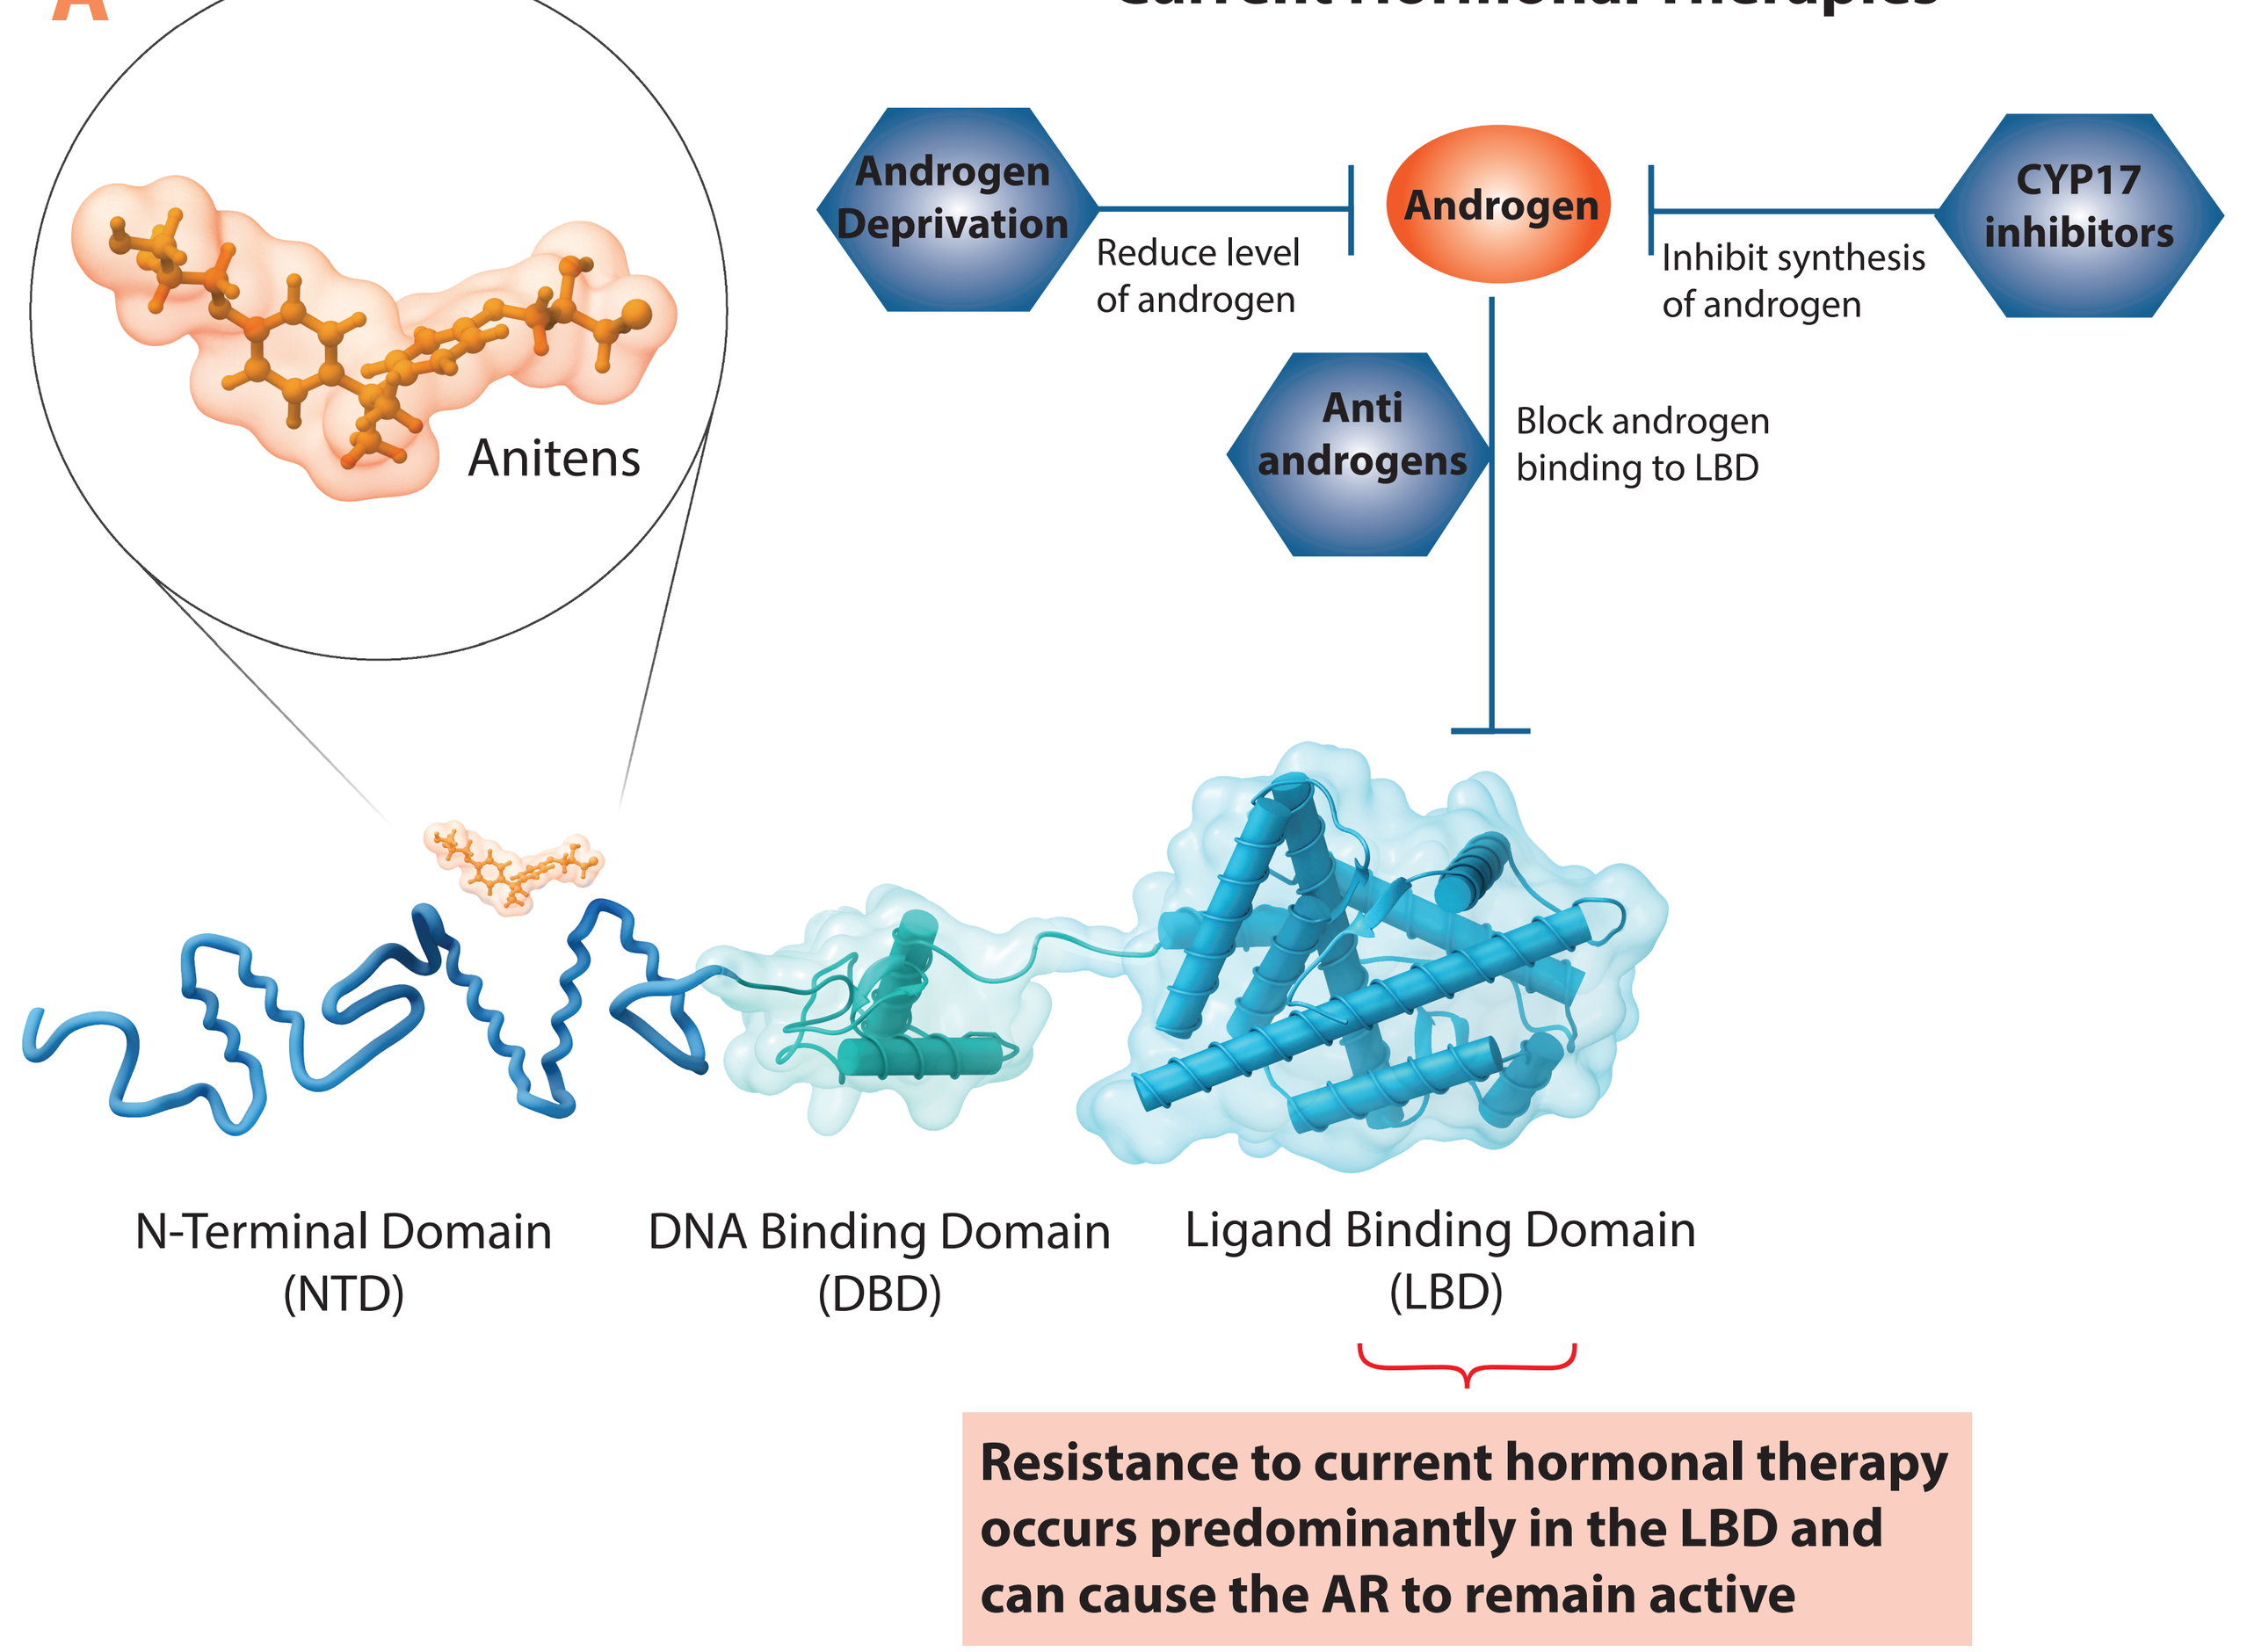
Current Hormonal Therapies
Androgen Deprivation
Reduce level of androgen
Androgen
Inhibit synthesis of androgen
CYP17 inhibitors
Anti androgens
Block androgen binding to LBD
Anitens
N-Terminal Domain (NTD)
DNA Binding Domain (DBD)
Ligand Binding Domain (LBD)
Resistance to current hormonal therapy occurs predominantly in the LBD and can cause the AR to remain active

Figure 1: Anitens are first in class NTD inhibitors of the androgen receptor
(A) Structure of the androgen receptor (AR) and mechanism of inhibitions. The AR is organized in 3 distinct domains: the LBD, involved in binding with androgens, the DBD, and the NTD, which orchestrate the transactivation of the receptor. Inhibiting the NTD with Anitens allows the inactivation of the AR downstream of the LBD, and is an effective strategy to counteract the resistance mechanisms involving the LBD (mutations, splice variants).

Next generation Anitens demonstrate a 10-20 fold improvement on the inhibition of androgen-induced AR transcriptional activity

A

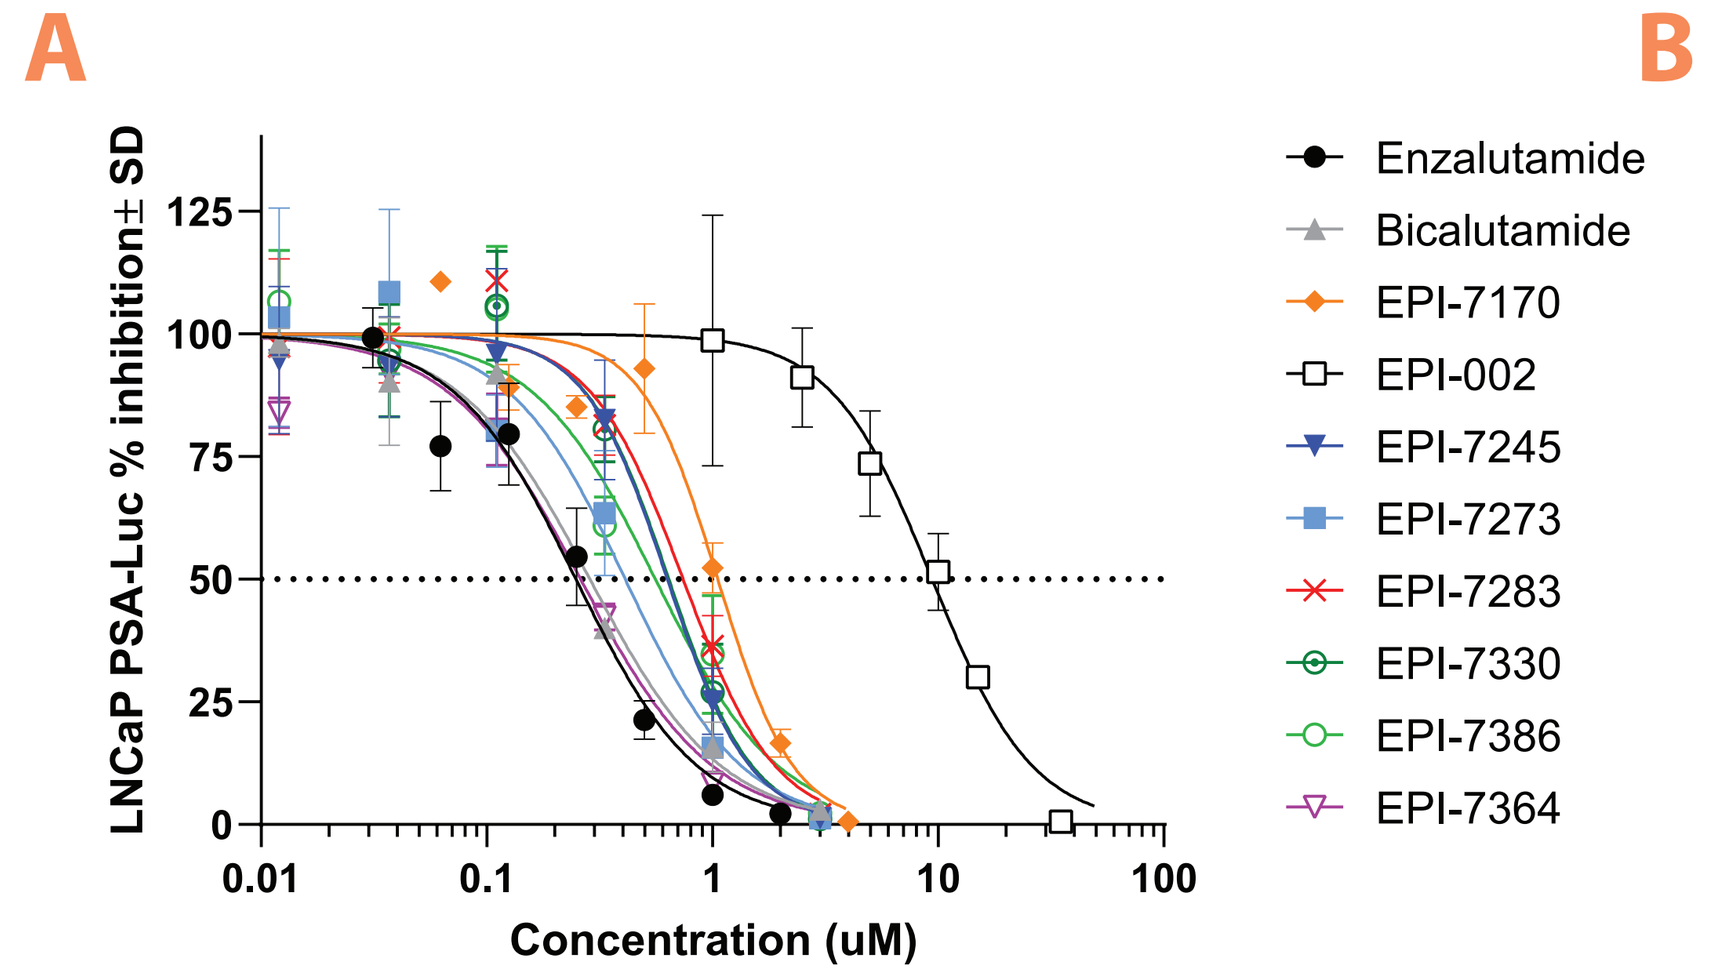
LNCaP PSA-Luc % inhibition± SD
Concentration (uM)
Enzalutamide
Bicalutamide
EPI-7170
EPI-002
EPI-7245
EPI-7273
EPI-7283
EPI-7330
EPI-7386
EPI-7364

B

Compound	IC50 (nM)	n
EPI-002	9,580	2
EPI-7170	1,054	3
EPI-7245	592	8
EPI-7273	400	5
EPI-7283	631	6
EPI-7330	658	6
EPI-7364	205	4
EPI-7386	535	2
Bicalutamide	306	2
Enzalutamide	189	8

Figure 2: Activity against androgen-induced PSA-luciferase activity in LNCaP cells
(A) A dose-dependent decrease in AR-transcriptional activity was demonstrated in LNCaP cells transfected with the PSA reporter gene and incubated with different Aniten compounds in the presence of androgen (R1881). (B) Summary of IC50s calculated across multiple independent experiments.

AR inhibition is on target, through the N-terminal domain and effective in AR-V7 driven models

A

	Steroid Receptor LBD IC50 (uM)				
Compound	AR	ER aplha	ER beta	PR	GR
EPI-002	>10	>10	>10	>10	>10
EPI-7170	9.38	5.82	7.37	4.96	>10
EPI-7245	>10	>10	>10	0.97	>10
EPI-7273	>10	>10	>10	0.93	>10
EPI-7283	>10	>10	>10	5.07	>10
EPI-7330	>10	>10	>10	5.45	9.51
EPI-7364	>10	>10	>10	5.04	>10
EPI-7386	>10	>10	>10	5.61	>10
Enzalutamide	0.54	>10	>10	4.49	>10

B

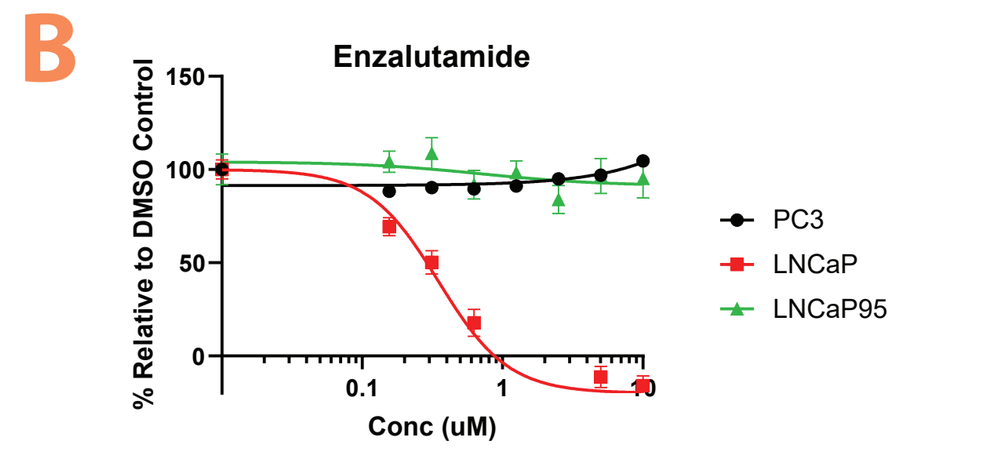
Enzalutamide
% Relative to DMSO Control
Conc (uM)
PC3
LNCaP
LNCaP95

C

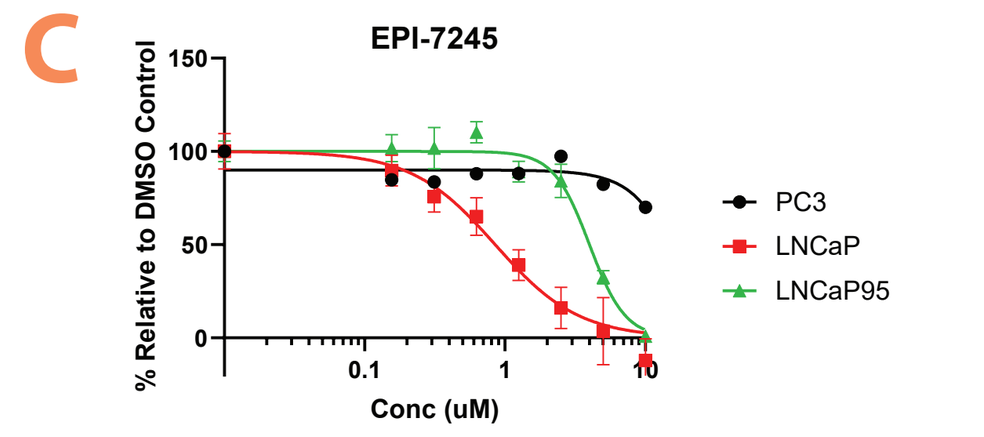
EPI-7245
% Relative to DMSO Control
Conc (uM)
PC3
LNCaP
LNCaP95

D

	Cellular proliferation IC50 (uM)		
Compound	LNCaP	PC-3	LNCaP95
EPI-002	9.00	>10	~20
EPI-7170	3.00	>10	4.00
EPI-7245	0.85	>10	4.00
EPI-7273	0.68	>10	6.43
EPI-7330	1.16	>10	2.65
EPI-7386	0.44	>10	3.78
Enzalutamide	0.35	>10	>10

E

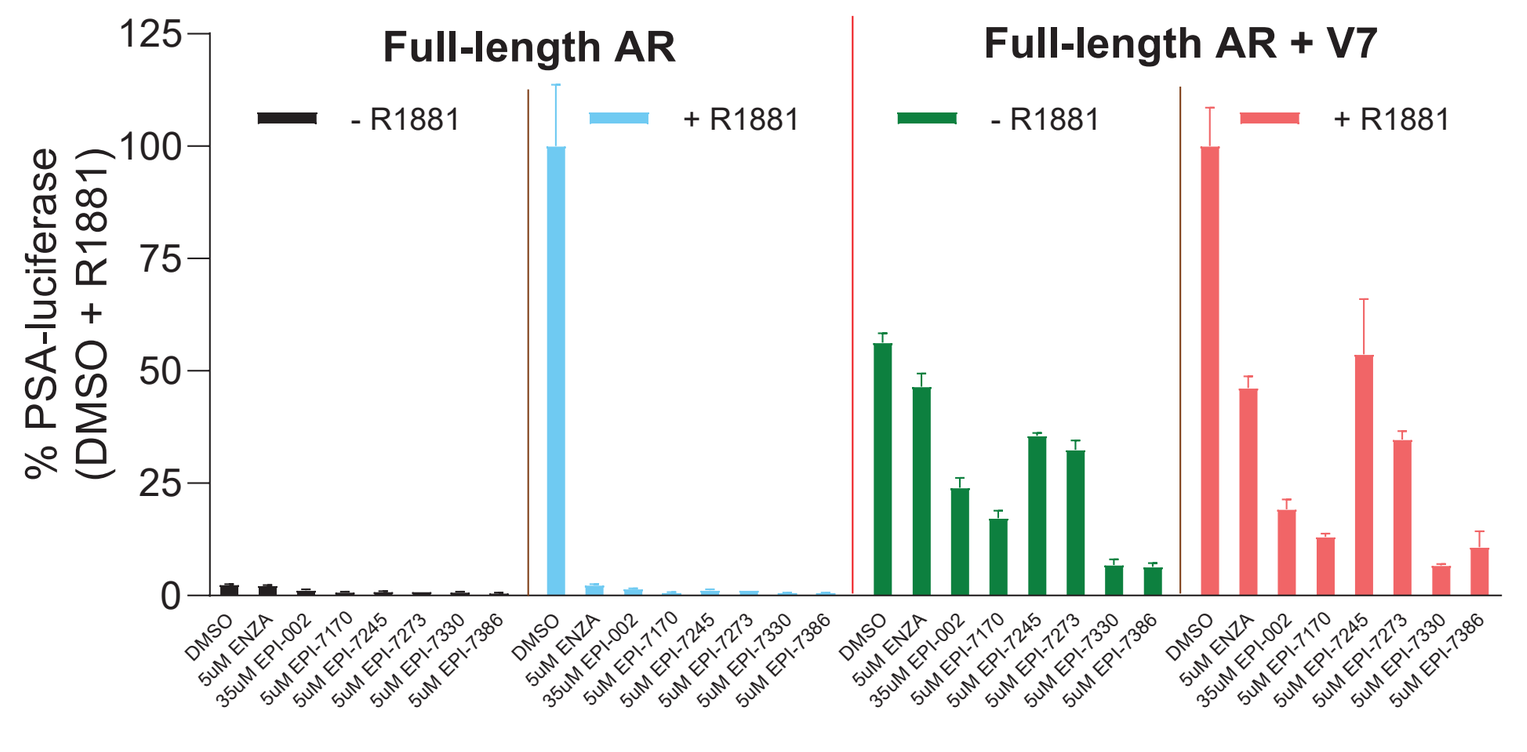
Full-length AR
Full-length AR + V7
- R1881
+ R1881
% PSA-luciferase (DMSO + R1881)

F

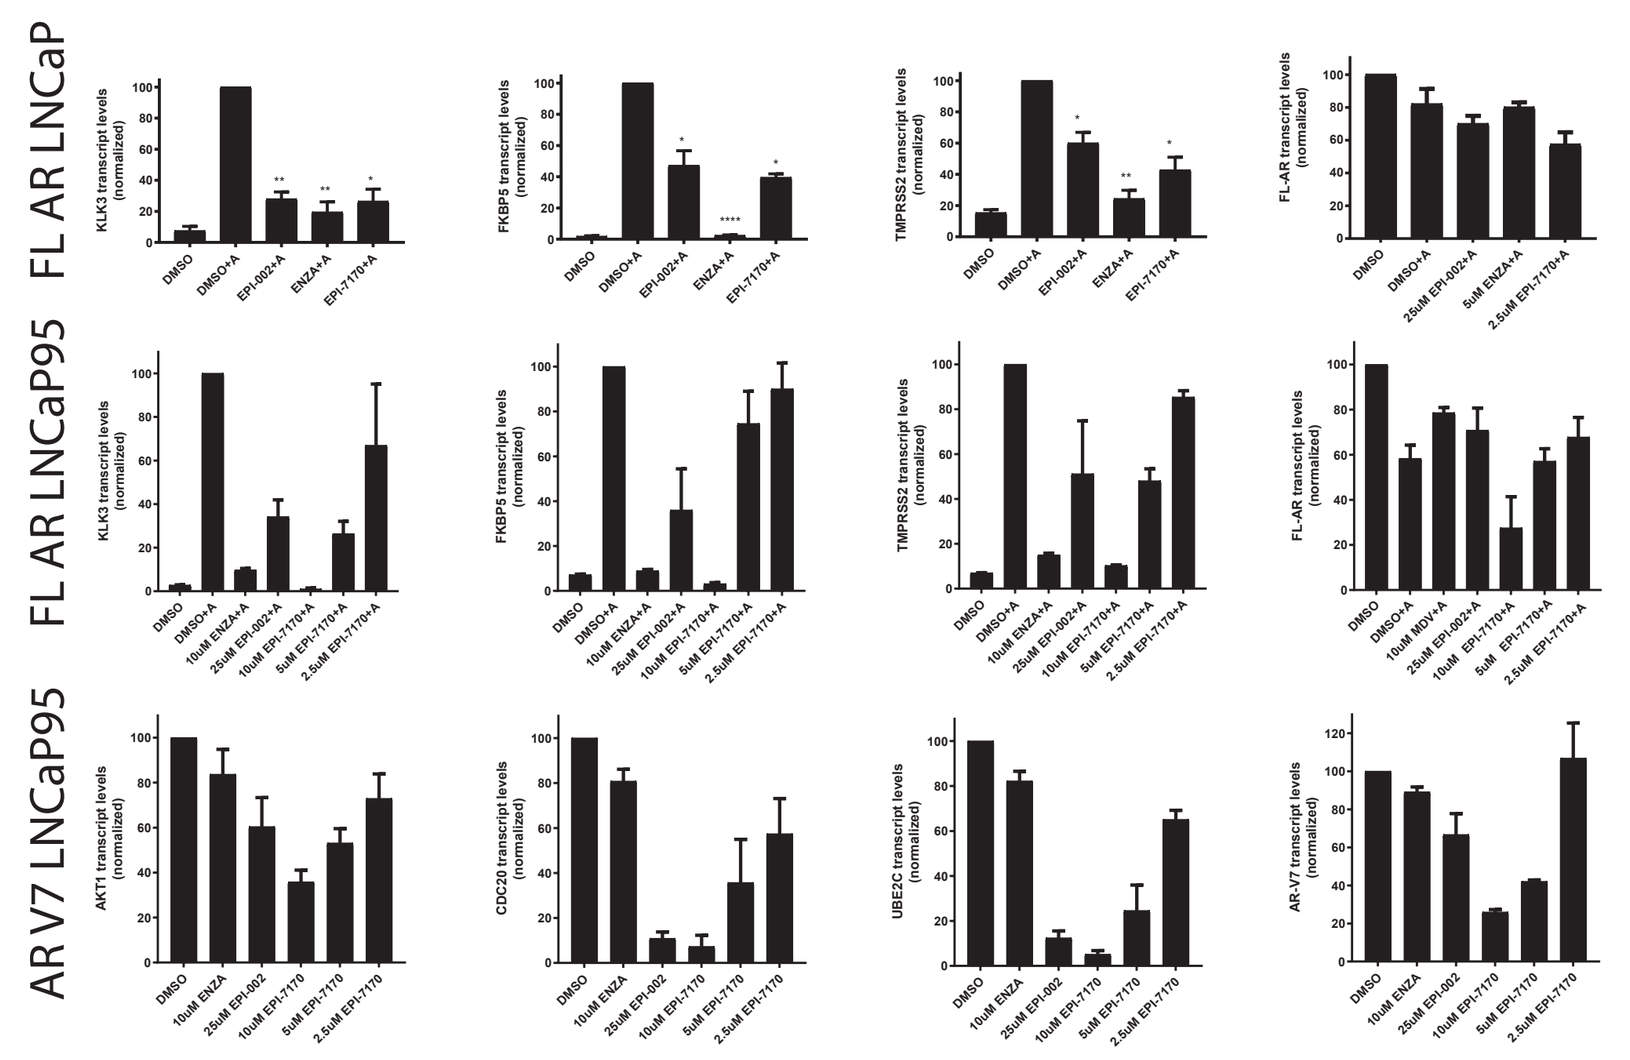
FL AR LNCaP
FL AR LNCaP95
AR V7 LNCaP95

Figure 3: Anitens activity and selectivity in AR WT and V7 models
(A) The selectivity of Anitens for AR NTD versus nuclear receptor LBDs was assessed using the GeneBLAzer assay (Thermo Fisher). The LBD-Gal4DBD fusion protein binds to UAS sites of the β-lactamase reporter to drive its expression. Estrogen receptor-α (ERα) and ERβ, progesterone receptor (PR), glucocorticoid receptor (GR). (B-D) The androgen-induced proliferation of LNCaP and viability of PC3 cells was measured with Alamar blue while BrdU was used for LNCaP95 cells. In LNCaP cells, AR specific proliferation was calculated by the difference between growth in the presence of androgen (0.1 nM R1881) versus in its absence. Enzalutamide (B), EPI-7245 (C) and summary table (D). (E) The transcriptional activities of endogenous full-length (FL) AR (+ R1881) and ectopic AR-V7 was measured in transiently transfected LNCaP cells using the PSA-luciferase reporter gene +/- R1881. (F) Endogenous expression of FL AR or V7-dependent target genes were measured by qPCR in either LNCaP (FL only) and LNCaP95 cells (FL and AR-V7) +/- R1881.

EPI-002 metabolic stability has been fixed with next generation Anitens, and is characterized in vivo with improved PK properties

A

	Liver microsome T1/2 (min)				
Compound	Human	Mouse	Rat	Dog	Monkey
EPI-002	>120	>120	N/A	N/A	N/A
EPI-7170	>120	N/A	N/A	N/A	N/A
EPI-7245	>120	>120	64	>120	>120
EPI-7273	>120	74	23	>120	73
EPI-7283	>120	>120	12	>120	>120
EPI-7330	>120	>120	>120	>120	>120
EPI-7364	>120	>120	83	>120	101
EPI-7386	>120	>120	>120	>120	>120
Enzalutamide	>120	>120	>120	>120	>120

B

	Hepatocytes T1/2 (min)		
Compound	Human	Mouse	Rat
EPI-002	109	63	N/A
EPI-7170	98	33	N/A
EPI-7245	290	203	75
EPI-7273	>360	86	31
EPI-7283	>360	>360	56
EPI-7330	>360	N/A	>360
EPI-7364	>360	>360	94
EPI-7386	>360	>360	>360
Enzalutamide	>360	N/A	N/A

C

Compound name	Route	Dose (mg/kg)	C0 (ng/mL)	T1/2 (h)	Vdss (L/kg)	Cl (mL/min/kg)	Tlast (h)	AUC0-last (ng.h/mL)	AUC0-inf (ng.h/mL)	MRT0-last (h)	MRT0-inf (h)	AUC Extra (%)	AUMC Extra (%)
EPI-7245	IV	0.50	395	5.1	3.10	7.67	18.7	1,010	1,140	5.1	7.5	11.1	38.2
EPI-7330			1,622	10.2	0.60	0.67	24.0	10,111	12,787	8.6	14.9	21.0	54.1
EPI-7386			913	8.9	0.58	0.78	24.0	9,142	10,913	8.1	12.7	15.6	44.2
Enzalutamide			568	19.1	1.11	0.68	24.0	7,206	12,392	10.5	27.7	41.4	77.1

D

Compound name	Route	Dose (mg/kg)	Cmax (ng/mL)	Tmax (h)	T1/2 (h)	Tlast (h)	AUC0-last (ng·h/mL)	AUC0-inf (ng.h/mL)	MRT0-last (h)	MRT0-inf (h)	AUC Extra (%)	AUMC Extra (%)	%F
EPI-7245	PO	5.00	1,143	1.0	8.35	24.00	8,309	9,775	8	11.7	14.5	44.0	85.8
EPI-7330			6,307	2.7	12.48	24.00	72,711	97,398	9	17.2	24.5	59.6	71.9
EPI-7386			2,673	2.2	8.07	24.00	30,714	35,039	8	11.8	12.9	37.7	33.6
Enzalutamide			4,543	2.3	14.31	24.00	75,348	111,888	10	21.6	32.5	67.3	105.0

E

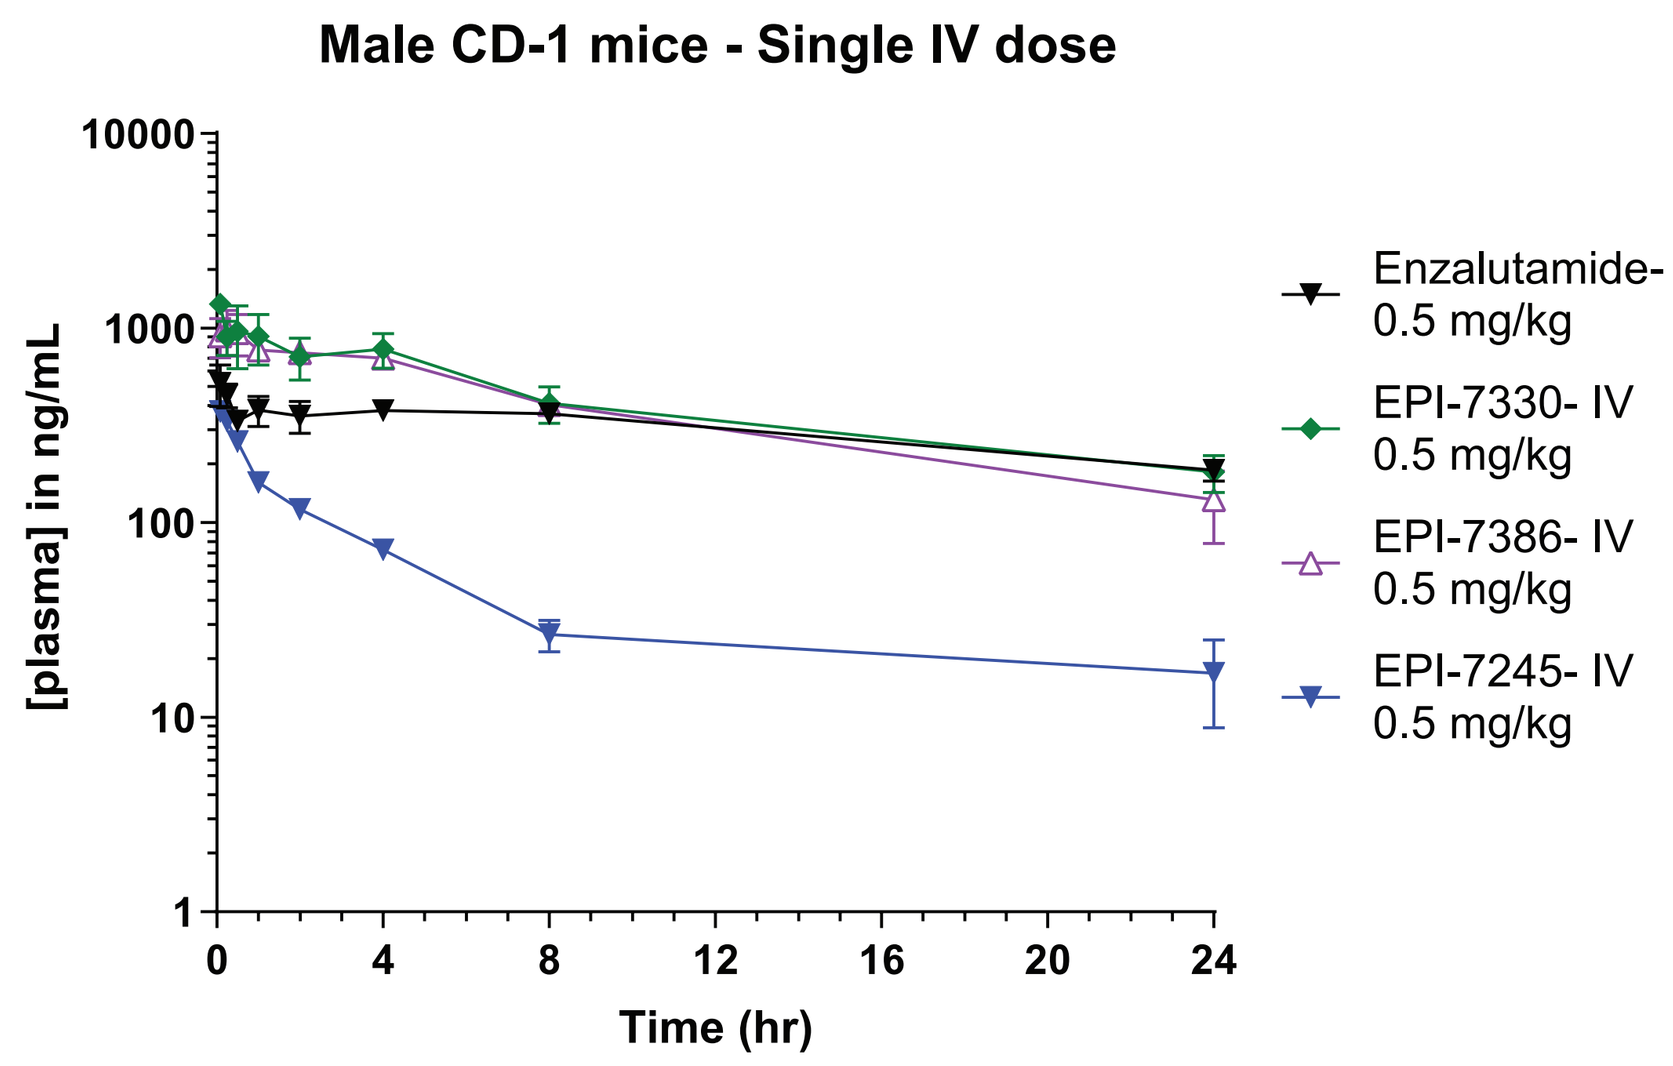
Male CD-1 mice - Single IV dose
[plasma] in ng/mL
Time (hr)
Enzalutamide- IV 0.5 mg/kg
EPI-7330- IV 0.5 mg/kg
EPI-7386- IV 0.5 mg/kg
EPI-7245- IV 0.5 mg/kg

F

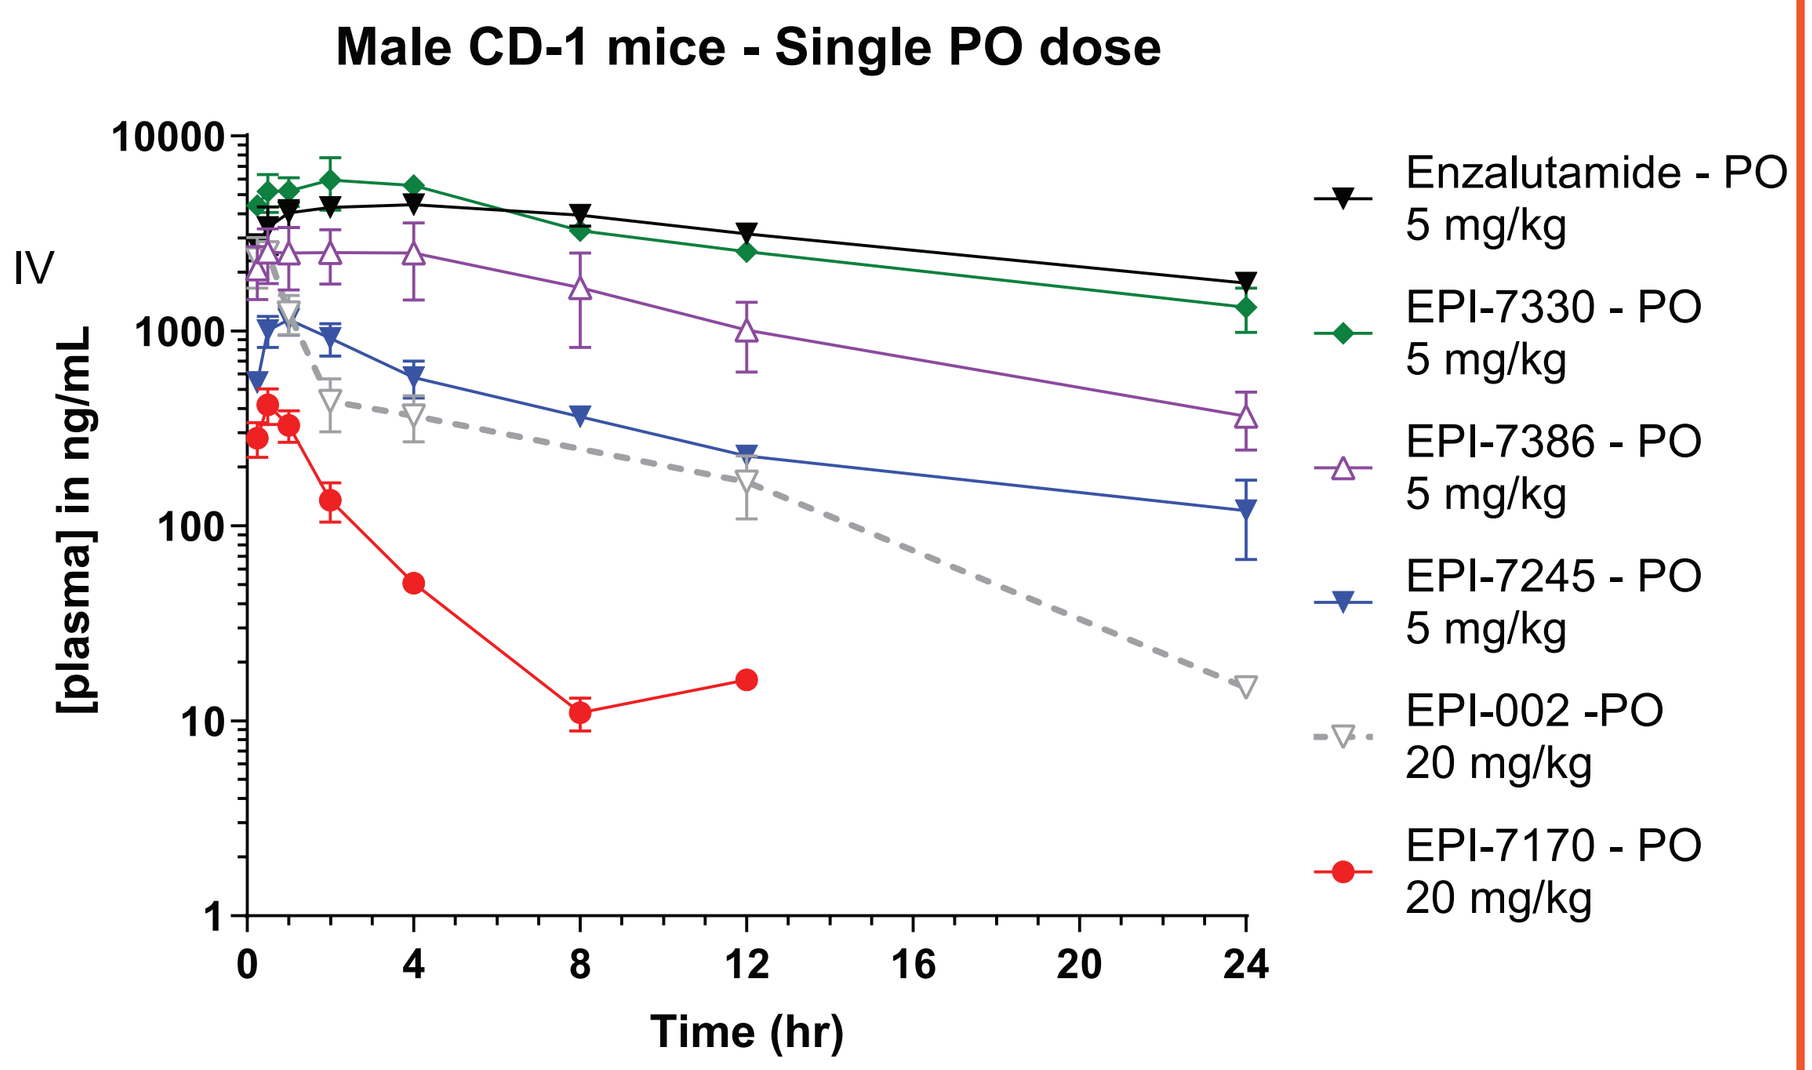
Male CD-1 mice - Single PO dose
[plasma] in ng/mL
Time (hr)
Enzalutamide - PO 5 mg/kg
EPI-7330 - PO 5 mg/kg
EPI-7386 - PO 5 mg/kg
EPI-7245 - PO 5 mg/kg
EPI-002 -PO 20 mg/kg
EPI-7170 - PO 20 mg/kg

G

Compound	Cmin at 5 mg/kg PO (uM)	Extrapolated Cmin in efficacy (uM)
EPI-7245	0.23	2.78
EPI-7330	2.42	29.1
EPI-7386	0.68	8.18
Enzalutamide	3.80	22.8

Figure 4: Anitens are metabolically stable and show adequate PK profile for high and sustained plasma exposure
Compound stability was assessed in human, mouse, rat, dog and monkey liver microsomes (A) and human and mouse hepatocytes (B). (C) Summary of IV PK parameters after a single dose in male CD-1 mice. (D) Summary of PO PK parameters after a single dose in male CD-1 mice. (E) PK curve after a single IV dose in male CD-1 mice. (F) PK curve after a single PO dose in CD-1 male mice. EPI-002 and EPI-7170 PO PK at 20 mg/kg were added as comparators. (G) Extrapolated drug concentration in plasma reached in the efficacy study at 60 mg/kg Aniten dosing, calculated based on single dose PK with no accumulation and dose linearity.

Next generation Anitens are active and well tolerated in castrated mice bearing AR dependent LNCaP xenografts

A

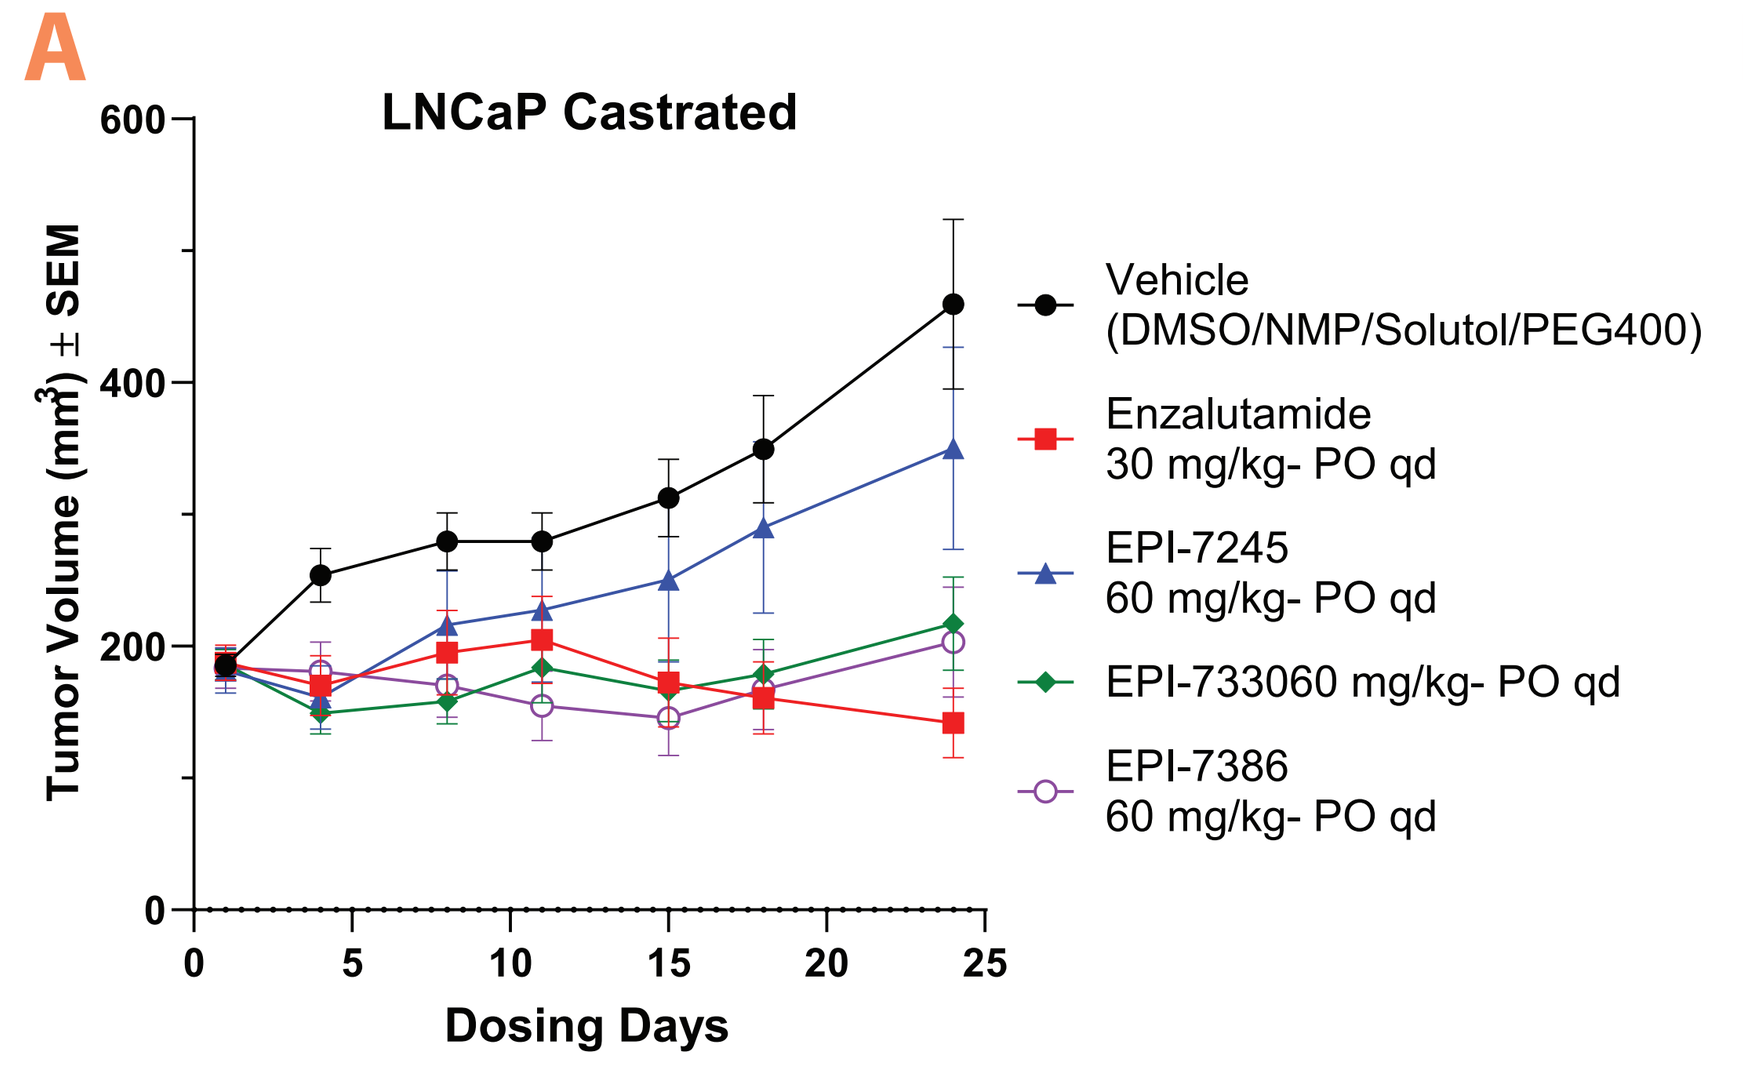
LNCaP Castrated
Tumor Volume (mm3) ± SEM
Dosing Days
Vehicle (DMSO/NMP/Solutol/PEG400)
Enzalutamide 30 mg/kg- PO qd
EPI-7245 60 mg/kg- PO qd
EPI-733060 mg/kg- PO qd
EPI-7386 60 mg/kg- PO qd

B

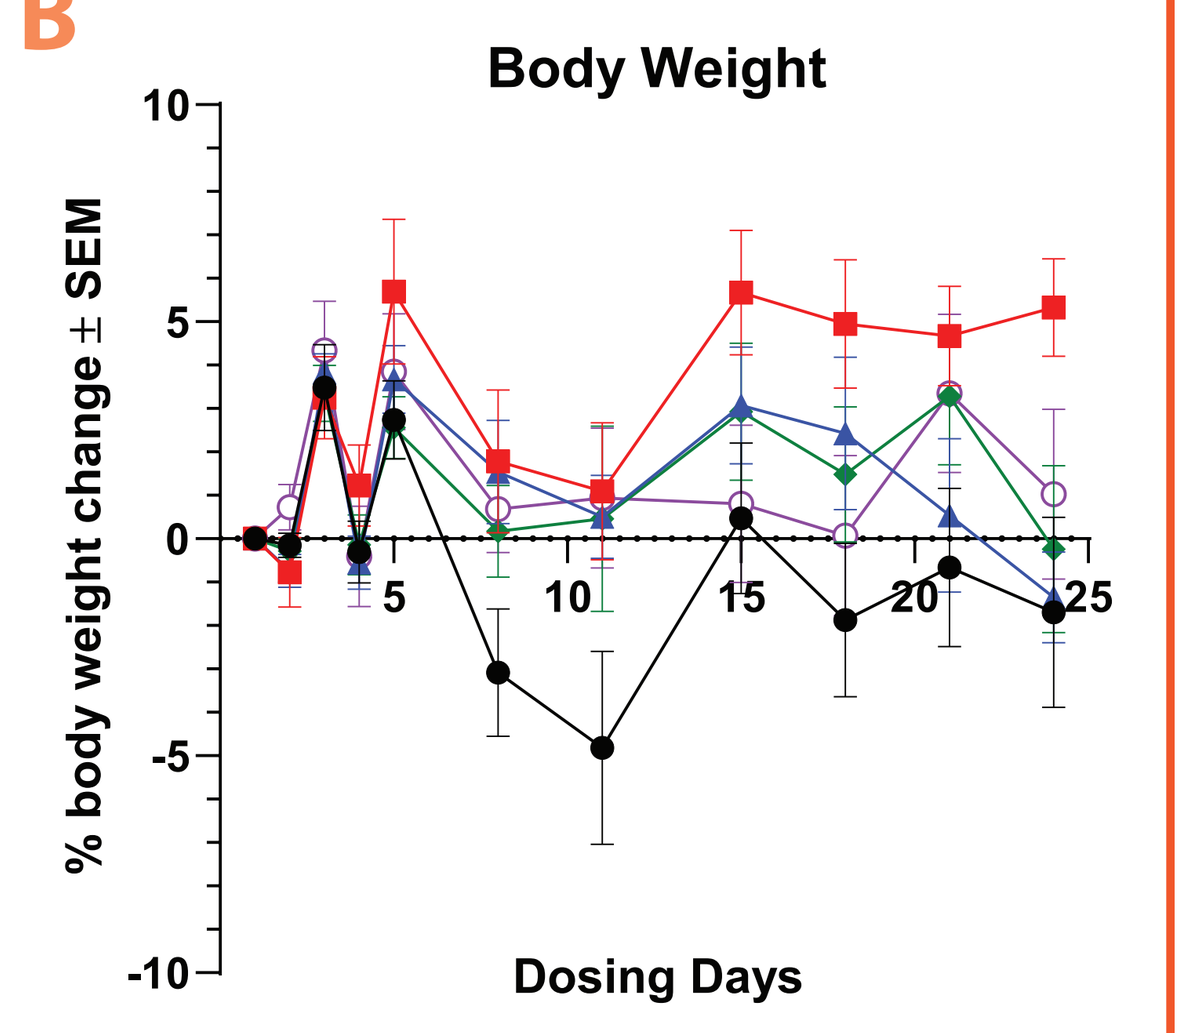
Body Weight
% body weight change ± SEM
Dosing Days

C

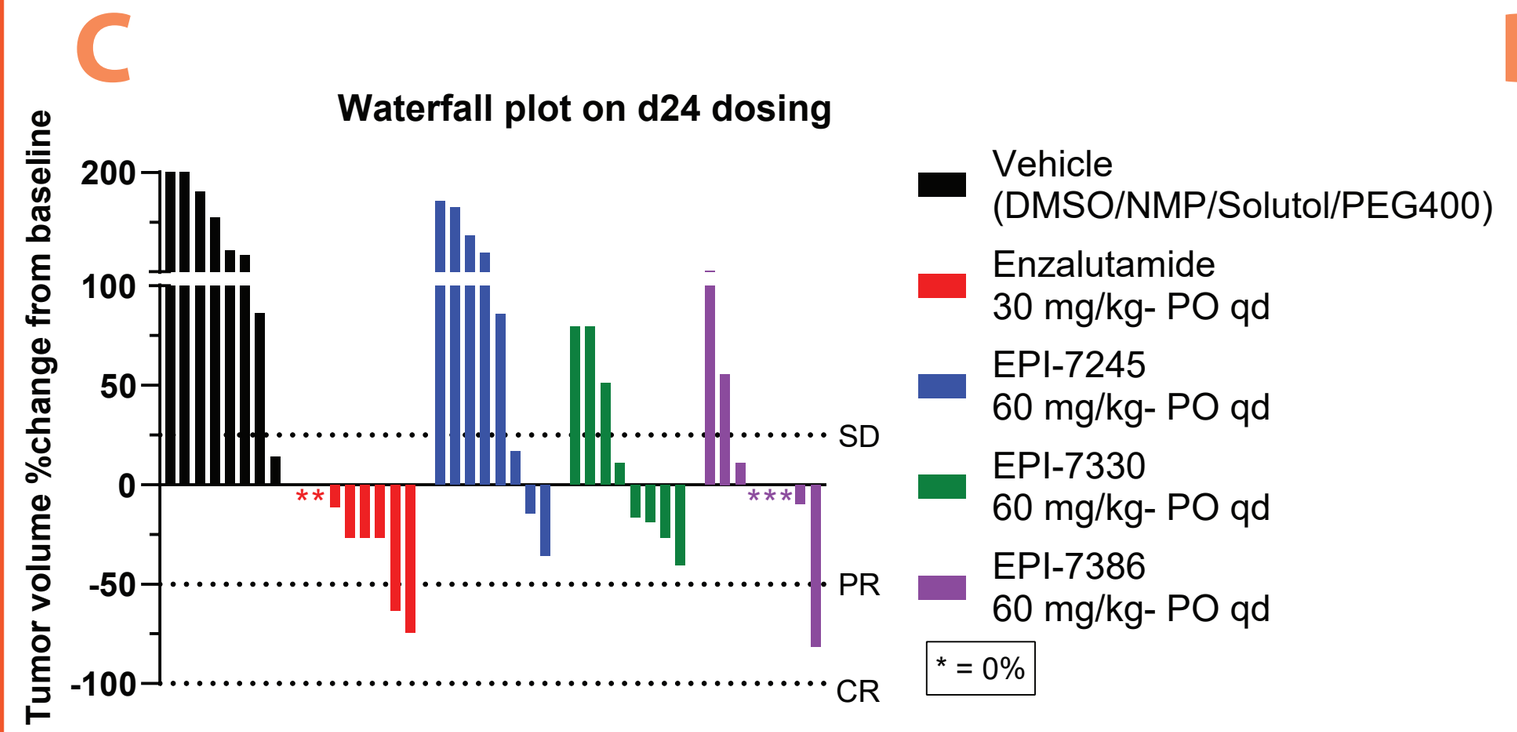
Waterfall plot on d24 dosing
Tumor volume %change from baseline
SD
PR
CR
Vehicle (DMSO/NMP/Solutol/PEG400)
Enzalutamide 30 mg/kg- PO qd
EPI-7245 60 mg/kg- PO qd
EPI-7330 60 mg/kg- PO qd
EPI-7386 60 mg/kg- PO qd
* = 0%

D

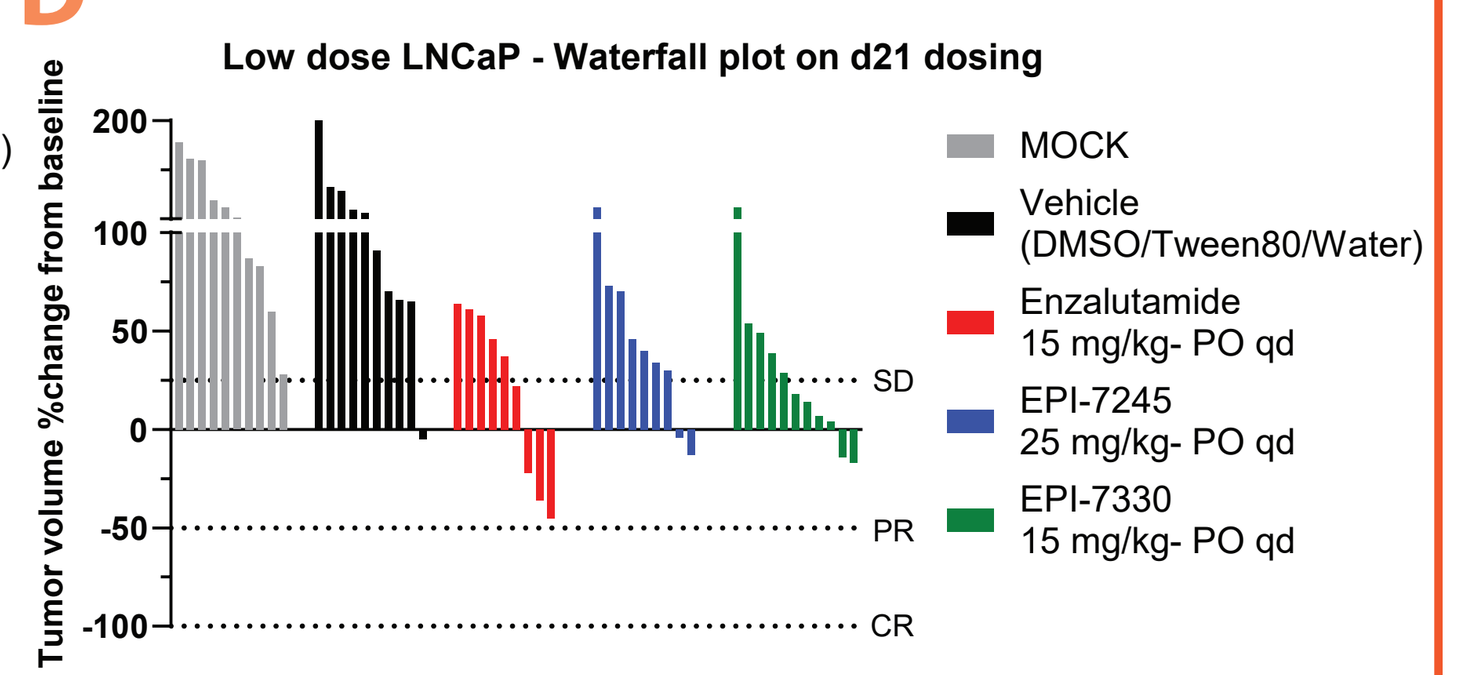
Low dose LNCaP - Waterfall plot on d21 dosing
Tumor volume %change from baseline
SD
PR
CR
MOCK
Vehicle (DMSO/Tween80/Water)
Enzalutamide 15 mg/kg- PO qd
EPI-7245 25 mg/kg- PO qd
EPI-7330 15 mg/kg- PO qd

Figure 5: In vivo activity in the LNCaP CRPC xenograft model
(A) Tumor growth measured in male NCG mice bearing LNCaP tumors. Castration was performed when tumors reached ~ 100 mm³ and dosing started 1 week after castration. (B) Body weight captured biweekly in the animals showed no drug related toxicity. (C) Individual tumor volume change from baseline measured at the end of the experiment. (D) Individual tumor volume change from baseline measured in a repeat LNCaP castrated xenograft done with lower doses of Anitens and a clinically relevant dose of Enzalutamide.

CONCLUSION

- Promising next-generation Aniten compounds have been identified
- Major chemistry efforts led to the identification of several Anitens with >10-20 fold improvement in cellular potency compared to EPI-506 and which are also metabolically stable
- These compounds showed in vivo activity in an AR full length model, but also in an enzalutamide resistant model driven by AR-V7
- IND-selection preclinical studies are underway on the most promising Aniten's with an IND submission planned shortly

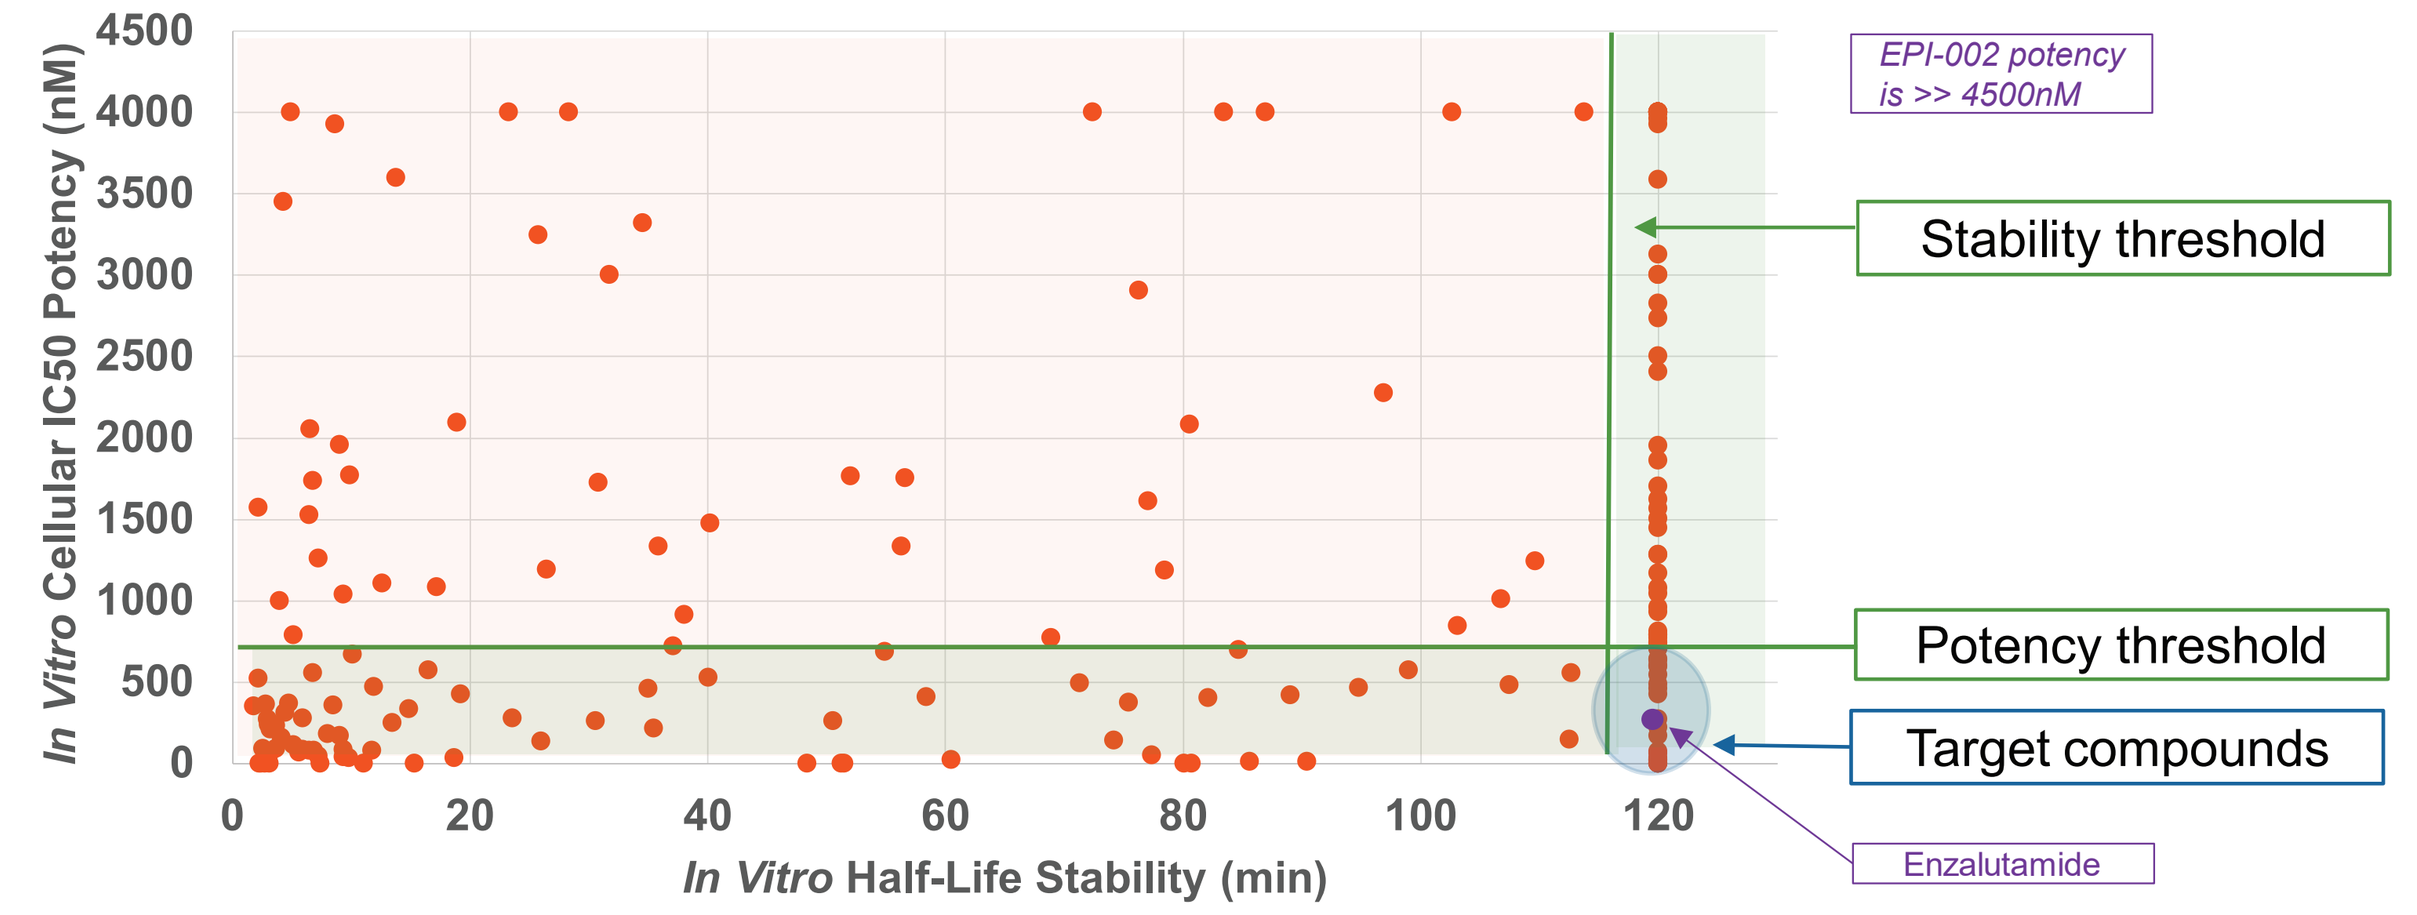
In Vitro Cellular IC50 Potency (nM)
In Vitro Half-Life Stability (min)
EPI-002 potency is >> 4500nM
Stability threshold
Potency threshold
Target compounds
Enzalutamide

Figure 6: Chemistry efforts led to the selection of potent and metabolically stable next generaton Anitens