A new generation of N-terminal domain androgen receptor inhibitors, with improved pharmaceutical properties, in castration resistant prostate cancer models

Abstract Number: 1292
Contact info:
rlemoigne@essapharma.com

Ronan Le Moigne[1], Nasrin R Mawji[2], C. Adriana Banuelos[2], Jun Wang[2], Kunzhong Jian[3], Raymond J. Andersen[3], Marianne D. Sadar[2], Han-Jie Zhou[1], Peter Virsik[1]

[1]ESSA Pharmaceuticals Inc., Houston, TX, and South San Francisco, USA, [2]Department of Genome Sciences Centre, BC Cancer Agency, 675 West 10th Avenue, Vancouver, BC V5Z 1L3, Canada, [3]Department of Chemistry, University of British Columbia, 2036 Main Mall, Vancouver, BC V6T 1Z1, Canada

BACKGROUND

The androgen receptor (AR) pathway continues to drive most castration-resistant prostate cancer (CRPC) even in late stages of the disease through resistance mechanisms including gain-of-function mutations in the C-terminal ligand-binding domain (LBD) and expression of constitutively active truncated AR splice variants lacking the LBD such as AR-V7. Selective inhibition of the N-terminal domain (NTD) of the AR can inhibit its transcriptional activity even in the presence of LBD-driven anti-androgen resistance.

A Phase I clinical trial of the first-generation AR NTD inhibitor, EPI-506, (a triacetate prodrug of EPI-002 - Ralaniten) demonstrated PSA declines in enzalutamide and/or abiraterone resistant metastatic CRPC patients. However, these declines were less than 50% and of short duration, revealing the need for more potent and metabolically stable NTD inhibitors.

A new generation of NTD transcriptional inhibitors (Anitens) has been generated. Examples of this new class, demonstrating improved potency, metabolic stability and pharmaceutical properties, are discussed in this poster.

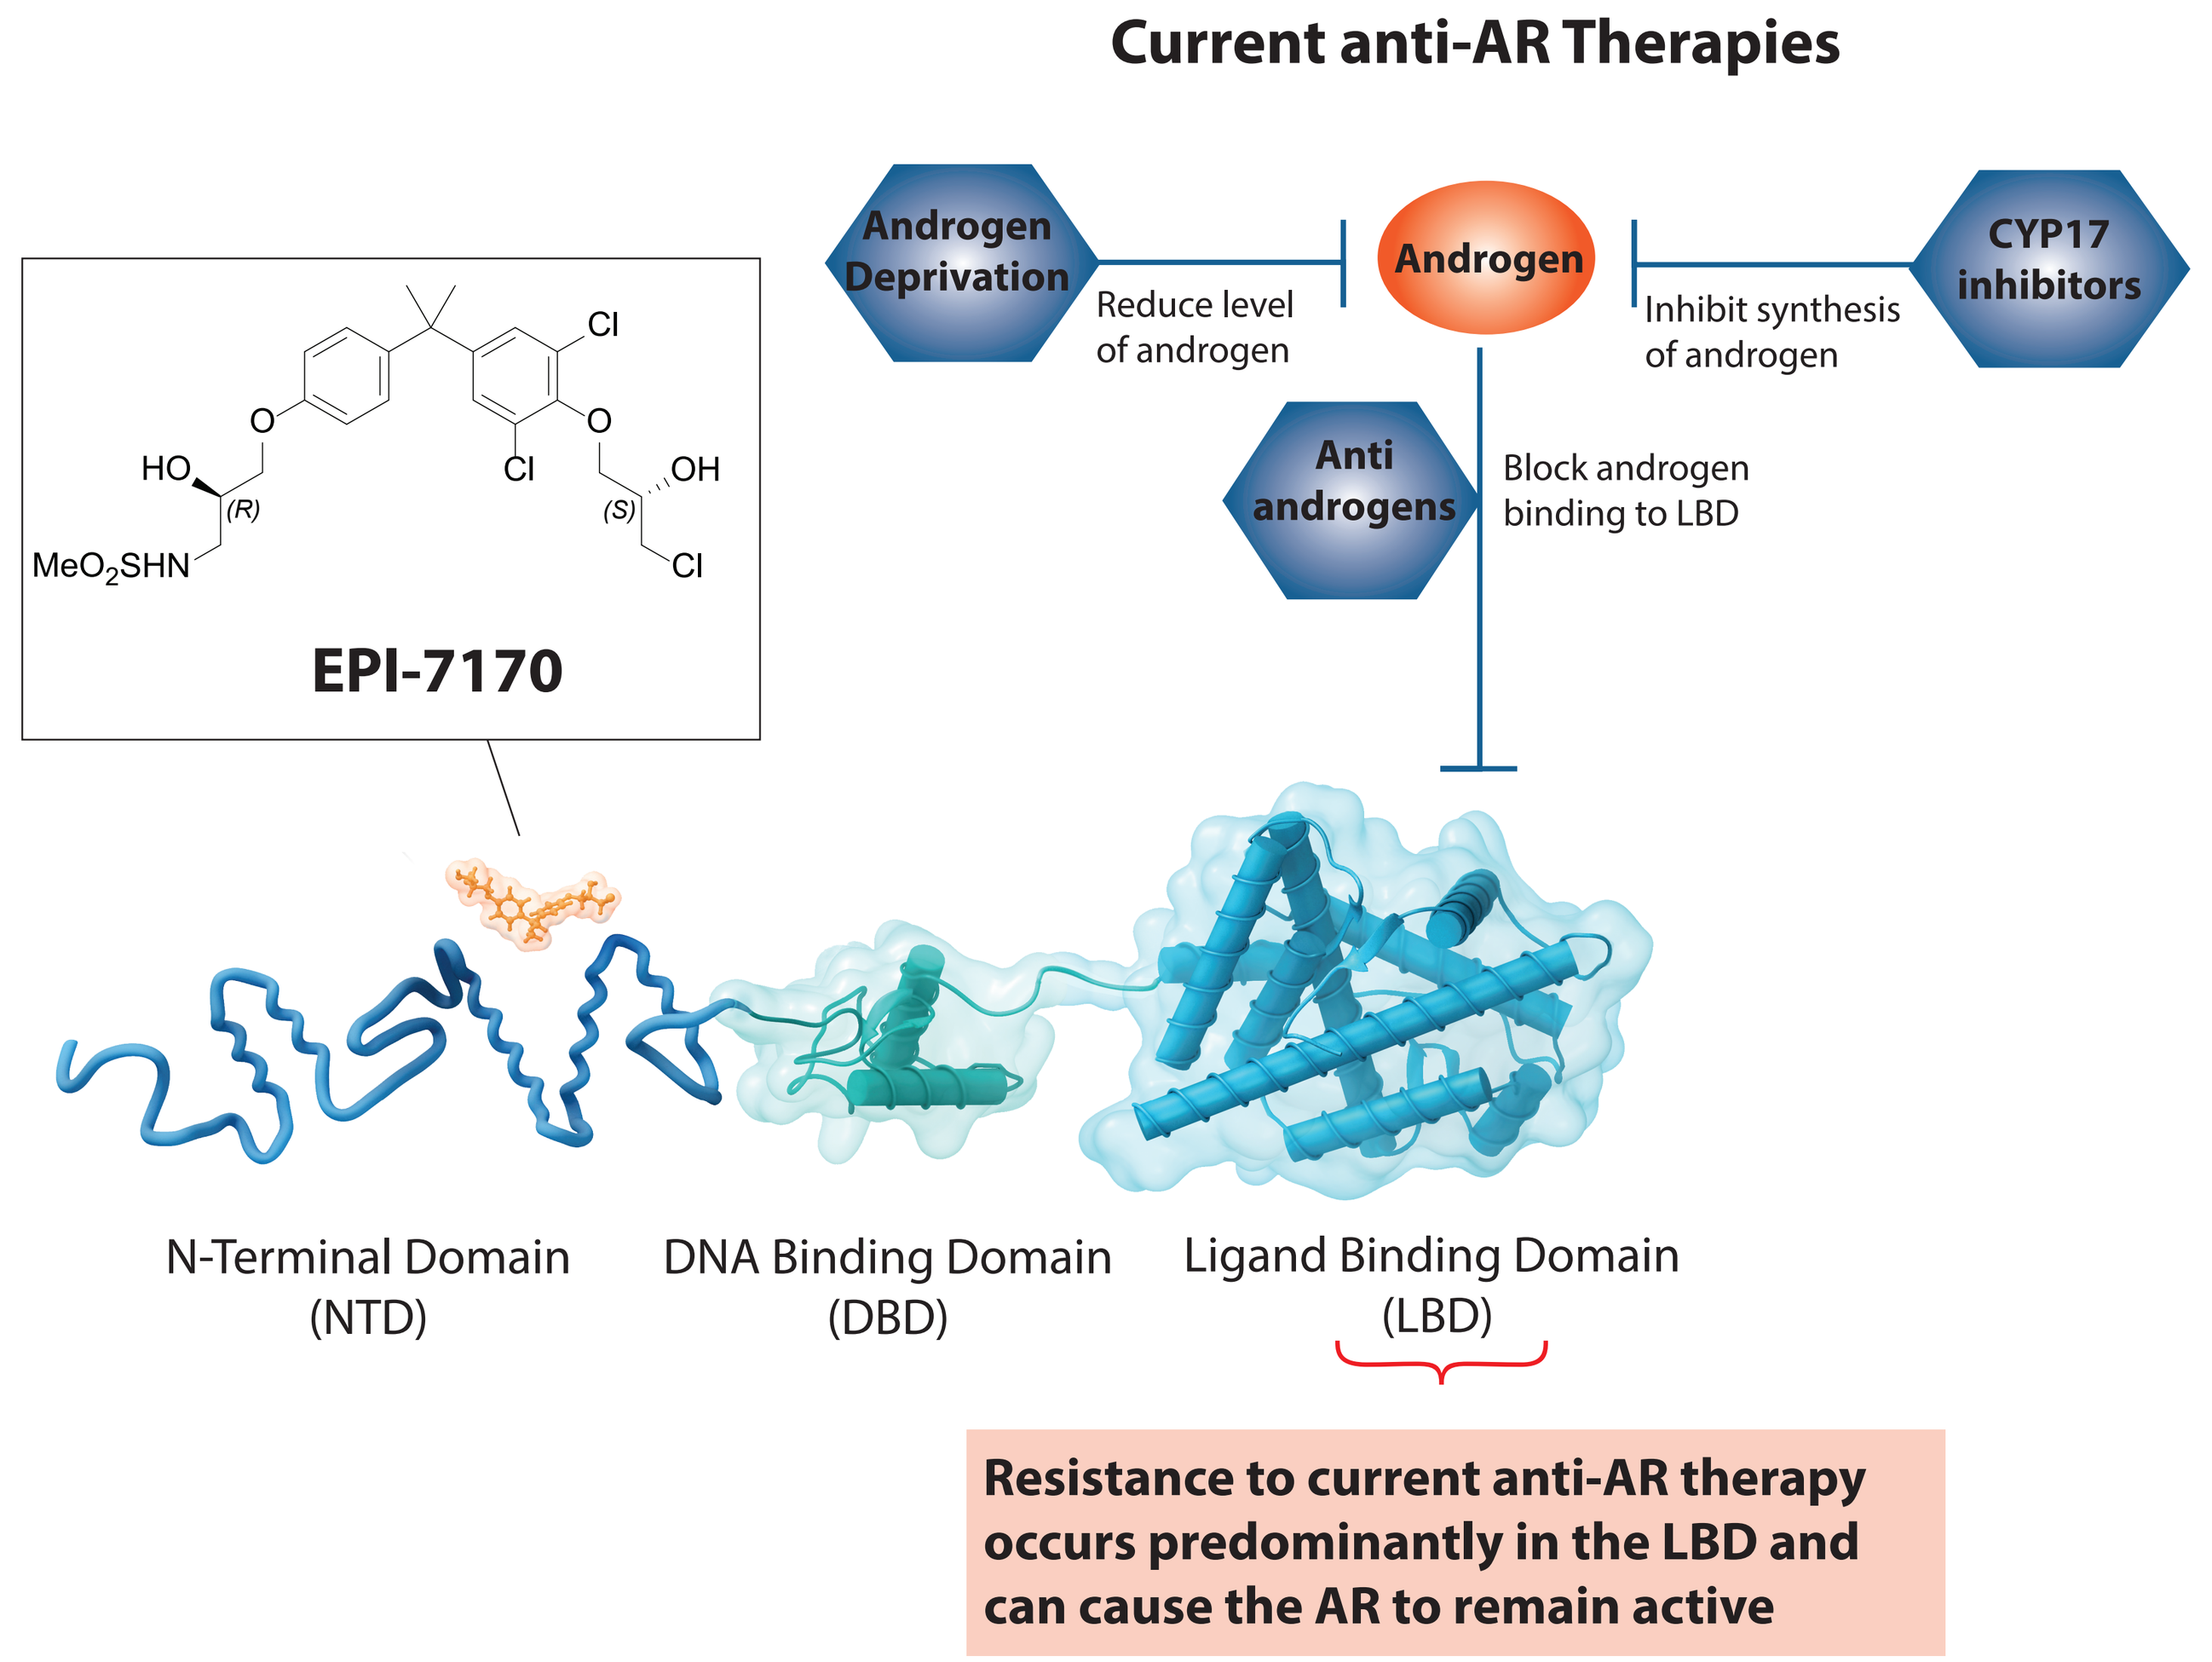

Current anti-AR Therapies
Androgen Deprivation
Reduce level of androgen
Androgen
Inhibit synthesis of androgen
CYP17 inhibitors
Anti androgens
Block androgen binding to LBD
EPI-7170
N-Terminal Domain (NTD)
DNA Binding Domain (DBD)
Ligand Binding Domain (LBD)
Resistance to current anti-AR therapy occurs predominantly in the LBD and can cause the AR to remain active

Figure 1: Anitens are first in class NTD inhibitors of the androgen receptor
Structure of the androgen receptor (AR) and mechanism of inhibitions. The AR is organized in 3 distinct domains: the LBD, involved in binding with androgens, the DBD, and the NTD, which orchestrate the transactivation of the receptor. Inhibiting the NTD with Anitens, exemplified with EPI-7170, allows the inactivation of the AR downstream of the LBD, and is an effective strategy to counteract the resistance mechanisms involving the LBD (mutations, splice variants).

Next generation Anitens demonstrate up to 20-fold improvement on the inhibition of androgen-induced AR transcriptional activity

A

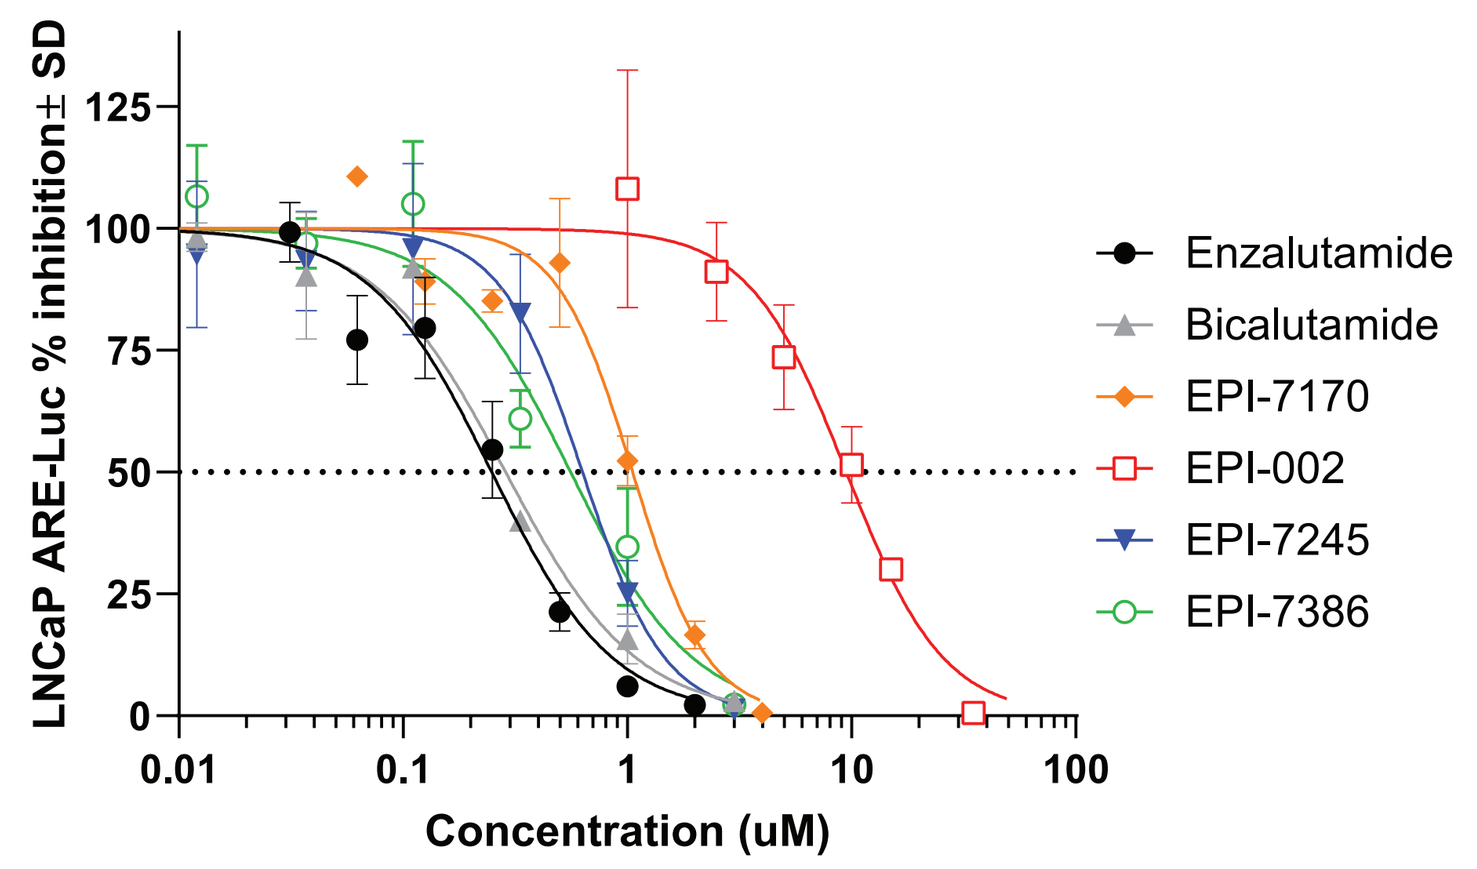

LNCaP ARE-Luc % inhibition± SD
Concentration (uM)
Enzalutamide
Bicalutamide
EPI-7170
EPI-002
EPI-7245
EPI-7386

B

Compound	IC50 (nM)	n
EPI-002	9,580	2
EPI-7170	1,054	3
EPI-7245	592	8
EPI-7386	513	3
Bicalutamide	306	2
Enzalutamide	189	8

Figure 2: Effect against androgen-induced PSA-luciferase activity in LNCaP cells
(A) A dose-dependent decrease in AR-transcriptional activity was demonstrated in LNCaP cells transfected with the PSA reporter gene and incubated with different Aniten compounds in the presence of androgen (R1881). (B) Summary of IC50s calculated across multiple independent experiments.

AR inhibition is on target, through the N-terminal domain, and effective in AR-V7 driven models

A

Compound	Steroid Receptor LBD inhibition IC50 (uM) AR	ER aplha	ER beta	PR	GR
EPI-002	>10	>10	>10	>10	>10
EPI-7170	9.38	5.82	7.37	4.96	>10
EPI-7245	>10	>10	>10	0.97	>10
EPI-7386	>10	>10	>10	5.61	>10
Enzalutamide	0.54	>10	>10	4.49	>10

B

Compound	PXR % activation at 10 uM	GABA Cl-channel (TBOB site) inhibition
EPI-002	17.6	21 uM IC50
EPI-7170	>66.5	68% at 10 uM
EPI-7245	ND	26% at 10 uM
EPI-7386	0.5	23% at 10 uM
Apalutamide	Inducer	3 uM IC50
Enzalutamide	Potential inducer	2.6 uM IC50

C

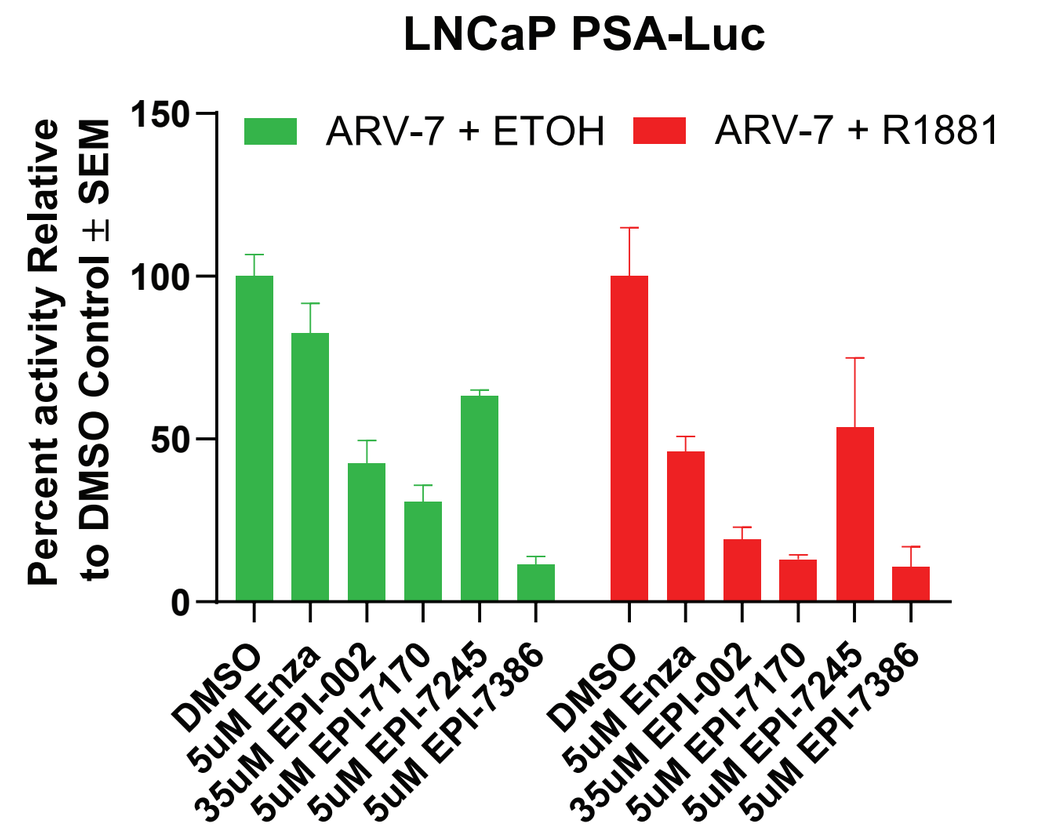

LNCaP PSA-Luc
ARV-7 + ETOH
ARV-7 + R1881
Percent activity Relative to DMSO Control ± SEM
DMSO
5uM Enza
35uM EPI-002
5uM EPI-7170
5uM EPI-7245
5uM EPI-7386

D

Compound	Cellular proliferation IC50 (uM) LNCaP	PC-3	LNCaP95
EPI-002	9.0	>10	~20
EPI-7170	2.8	11	4.4
EPI-7245	0.85	>10	4.0
EPI-7386	0.56	>10	3.7
Enzalutamide	0.35	>10	>10

E

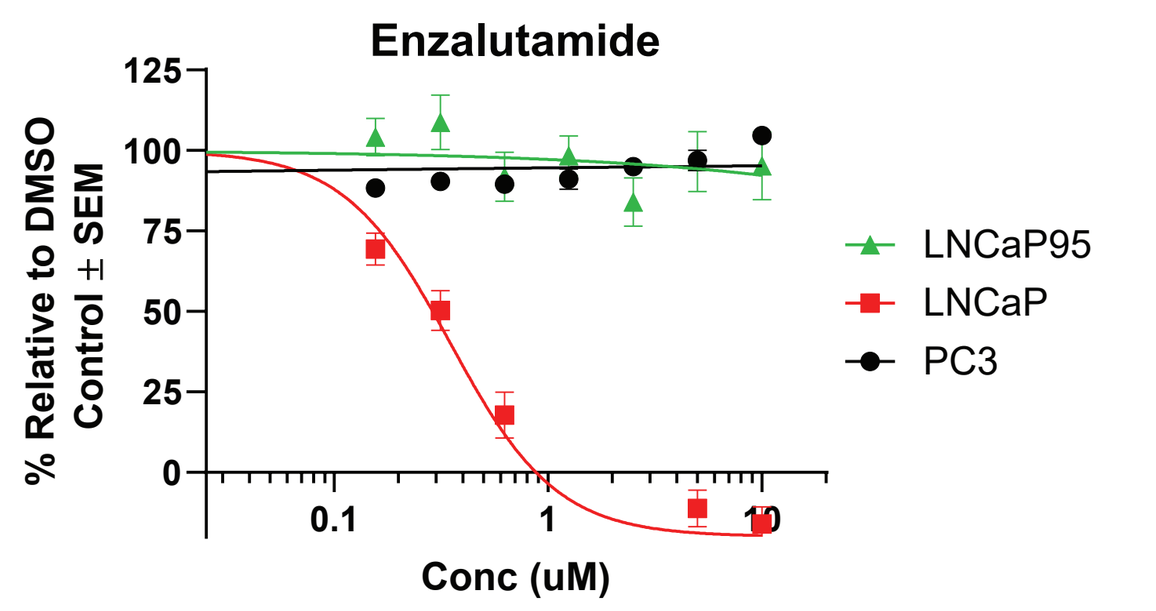

Enzalutamide
% Relative to DMSO Control ± SEM
Conc (uM)
LNCaP95
LNCaP
PC3

F

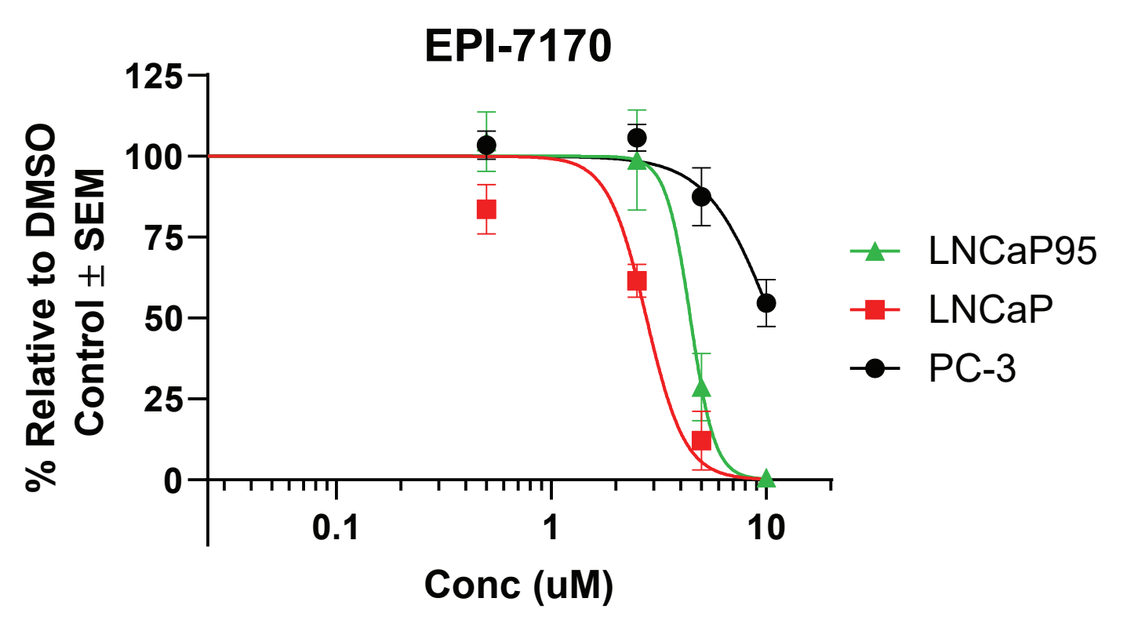

EPI-7170
% Relative to DMSO Control ± SEM
Conc (uM)
LNCaP95
LNCaP
PC-3

G

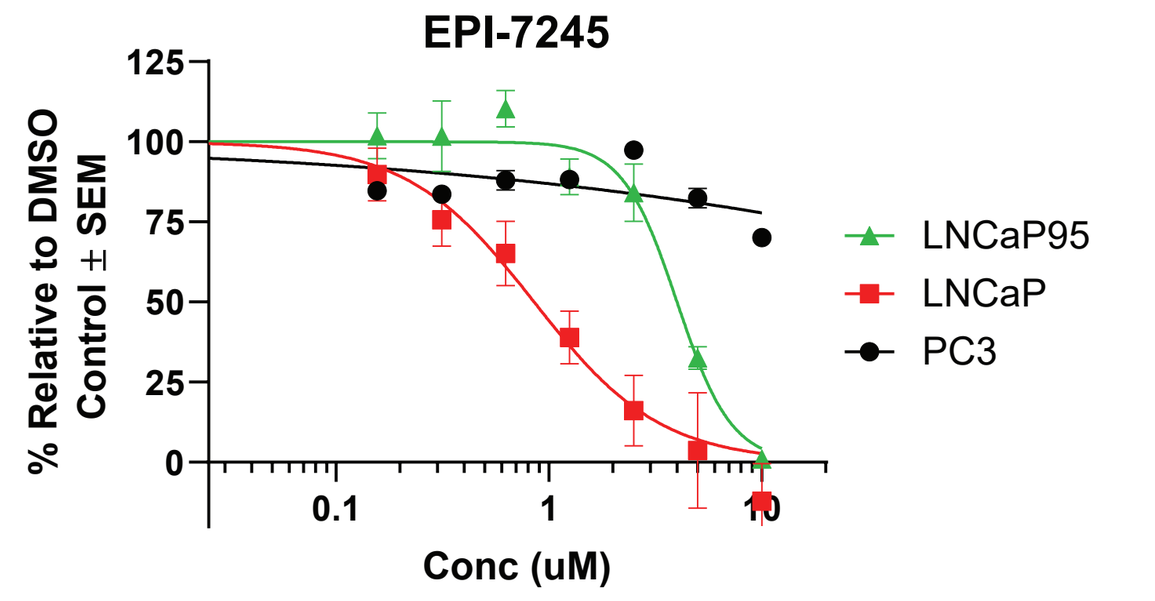

EPI-7245
% Relative to DMSO Control ± SEM
Conc (uM)
LNCaP95
LNCaP
PC3

H

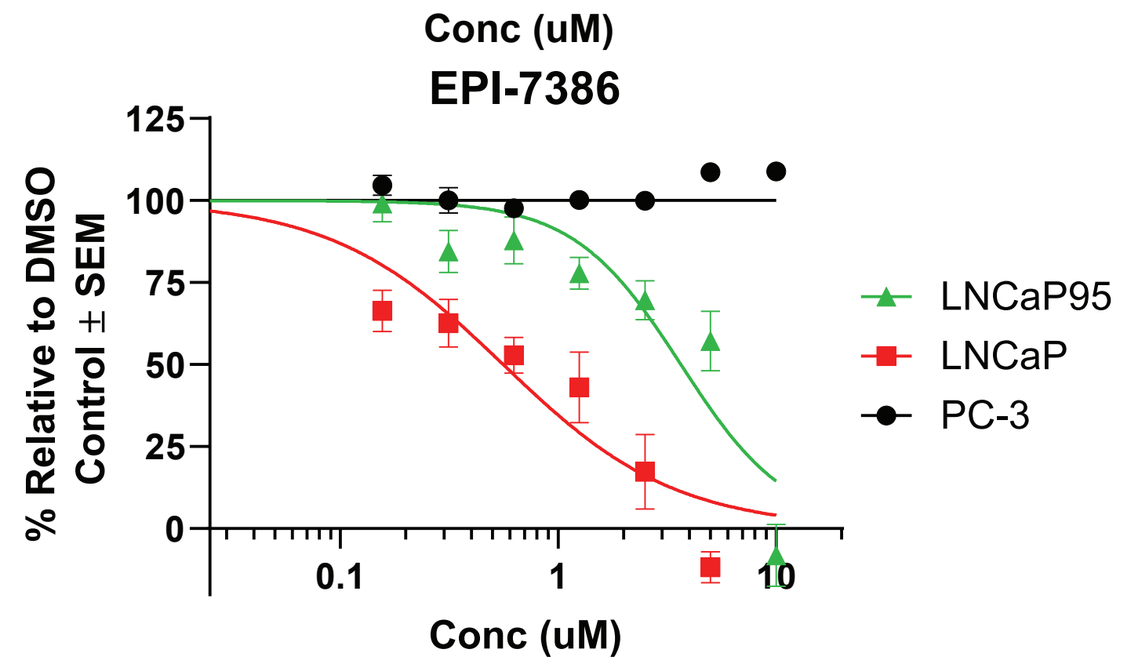

EPI-7386
% Relative to DMSO Control ± SEM
Conc (uM)
LNCaP95
LNCaP
PC-3

Figure 3: Anitens activity and selectivity in AR WT and V7 models
(A) The selectivity of Anitens for AR NTD versus other nuclear receptor LBDs was assessed using the GeneBLAzer assay (Thermo Fisher). Estrogen receptor-α (ERα) and ERβ, progesterone receptor (PR), glucocorticoid receptor (GR). (B) Pregnane X receptor (PXR) activity was measured using the DiscoverX functional assay while GABA-Cl channel antagonism was tested with CEREP binding assay. (C) The transcriptional activities of endogenous full-length (FL) AR (+ R1881) and ectopic AR-V7 was measured in transfected LNCaP cells using the PSA-luciferase reporter gene +/- R1881. (D-H) Androgen-induced proliferation of LNCaP and viability of PC3 cells was measured with Alamar blue while BrdU was used for LNCaP95 cells. Data is summarized in table D and represented in graphs E-H.

Next generation Anitens are metabolically stable and have high permeability, resulting in improved PK properties across species

A

Compound	Hepatocyte T1/2 (min) Human	Mouse	Rat
EPI-002	38	ND	ND
EPI-7170	98	33	ND
EPI-7245	290	203	64
EPI-7386	>360	>360	>360
Enzalutamide	>360	ND	ND

B

Compound ID	Mean Papp (10-6 cm/s) A to B	B to A	Efflux Ratio	Rank Papp	Efflux Transporter Substrate
Digoxin	0.04	13.87	332.3	Low	Likely
EPI-7386	4.14	5.5	1.3	High	Unlikely

Mouse PK

C

Compound name	Route	Dose (mg/kg)	C0 (ng/mL)	T1/2 (h)	Vdss (L/kg)	Cl (mL/min/kg)	AUC0-last (ng.h/mL)	AUC0-inf (ng.h/mL)	MRT0-last (h)
EPI-7245	IV	0.5	395	5.1	3.10	7.67	1,010	1,140	5.1
EPI-7386			913	8.9	0.58	0.78	9,142	10,913	8.1
EPI-002			693	6.5	4.00	13.55	493	711	1.5
Enzalutamide			568	19.1	1.11	0.68	7,206	12,392	10.5

D

Compound name	Route	Dose (mg/kg)	Cmax (ng/mL)	Tmax (h)	T1/2 (h)	AUC0-last (ng·h/mL)	AUC0-inf (ng.h/mL)	MRT0-last (h)	%F
EPI-7245	PO	5.0	1,143	1.0	8.35	8,309	9,775	8	85.8
EPI-7386			2,673	2.2	8.07	30,715	35,039	8	33.6
EPI-002			3,050	0.3	3.35	6,677	7,238	3	135.4
Enzalutamide			4,543	2.3	14.31	75,349	111,888	10	105.0

Dog PK

E

Compound name	Route	Dose (mg/kg)	C0 (ng/mL)	T1/2 (h)	Vdss (L/kg)	Cl (mL/min/kg)	AUC0-last (ng.h/mL)	AUC0-inf (ng.h/mL)	MRT0-last (h)
EPI-7386	IV	0.5	1,495	13.4	0.74	0.67	9,140	13,028	8.9

F

Compound name	Route	Dose (mg/kg)	Cmax (ng/mL)	Tmax (h)	T1/2 (h)	AUC0-last (ng·h/mL)	AUC0-inf (ng.h/mL)	MRT0-last (h)	%F
EPI-7386	PO	5.0	10,537	2.2	15.47	143,383	225,560	10	156.9

G

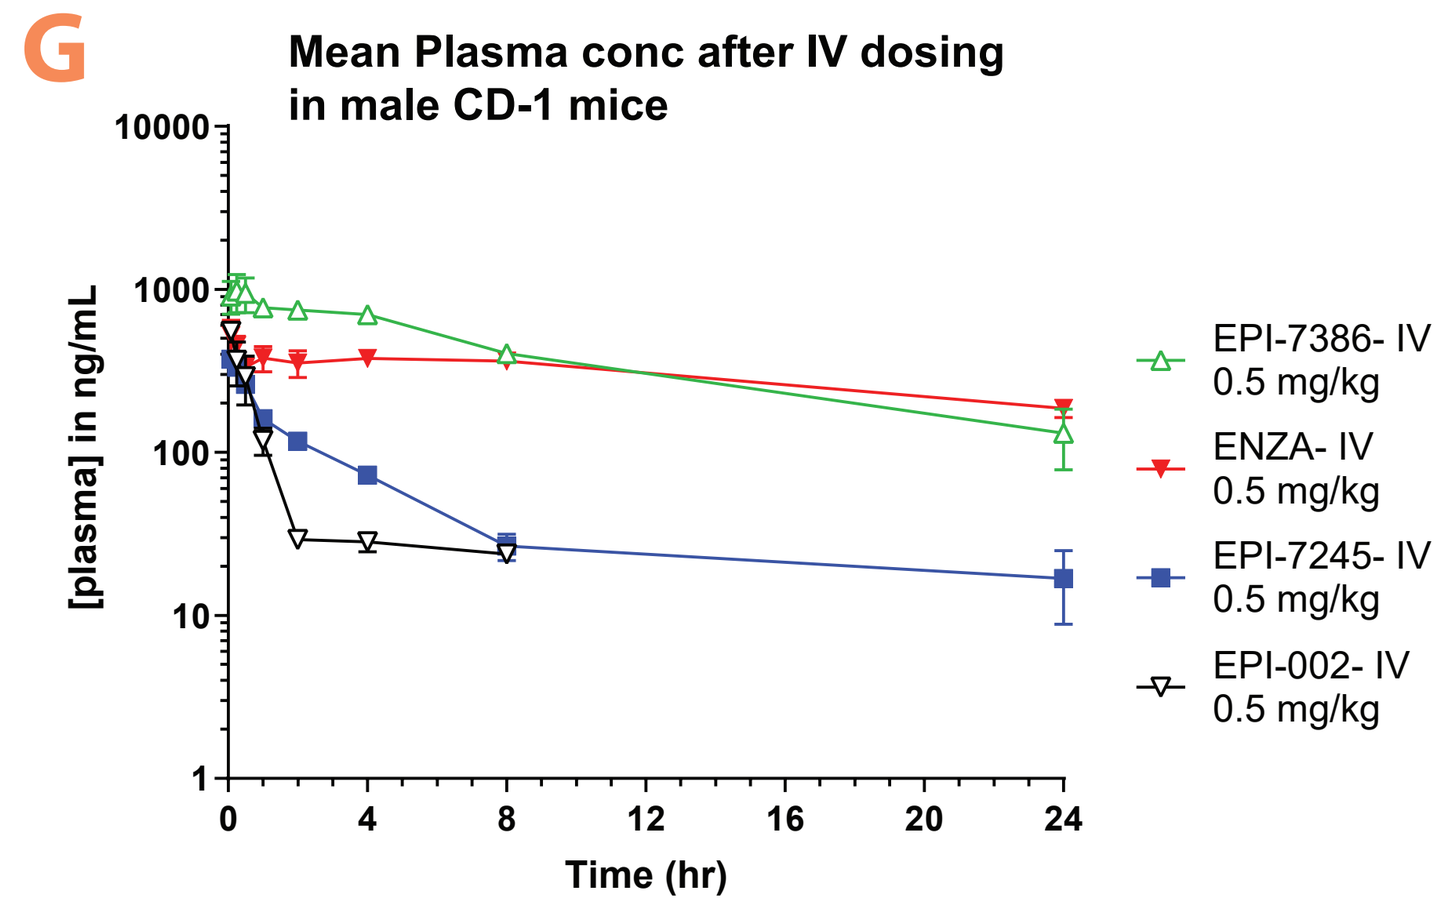

Mean Plasma conc after IV dosing in male CD-1 mice
[plasma] in ng/mL
Time (hr)
EPI-7386- IV 0.5 mg/kg
ENZA- IV 0.5 mg/kg
EPI-7245- IV 0.5 mg/kg
EPI-002- IV 0.5 mg/kg

H

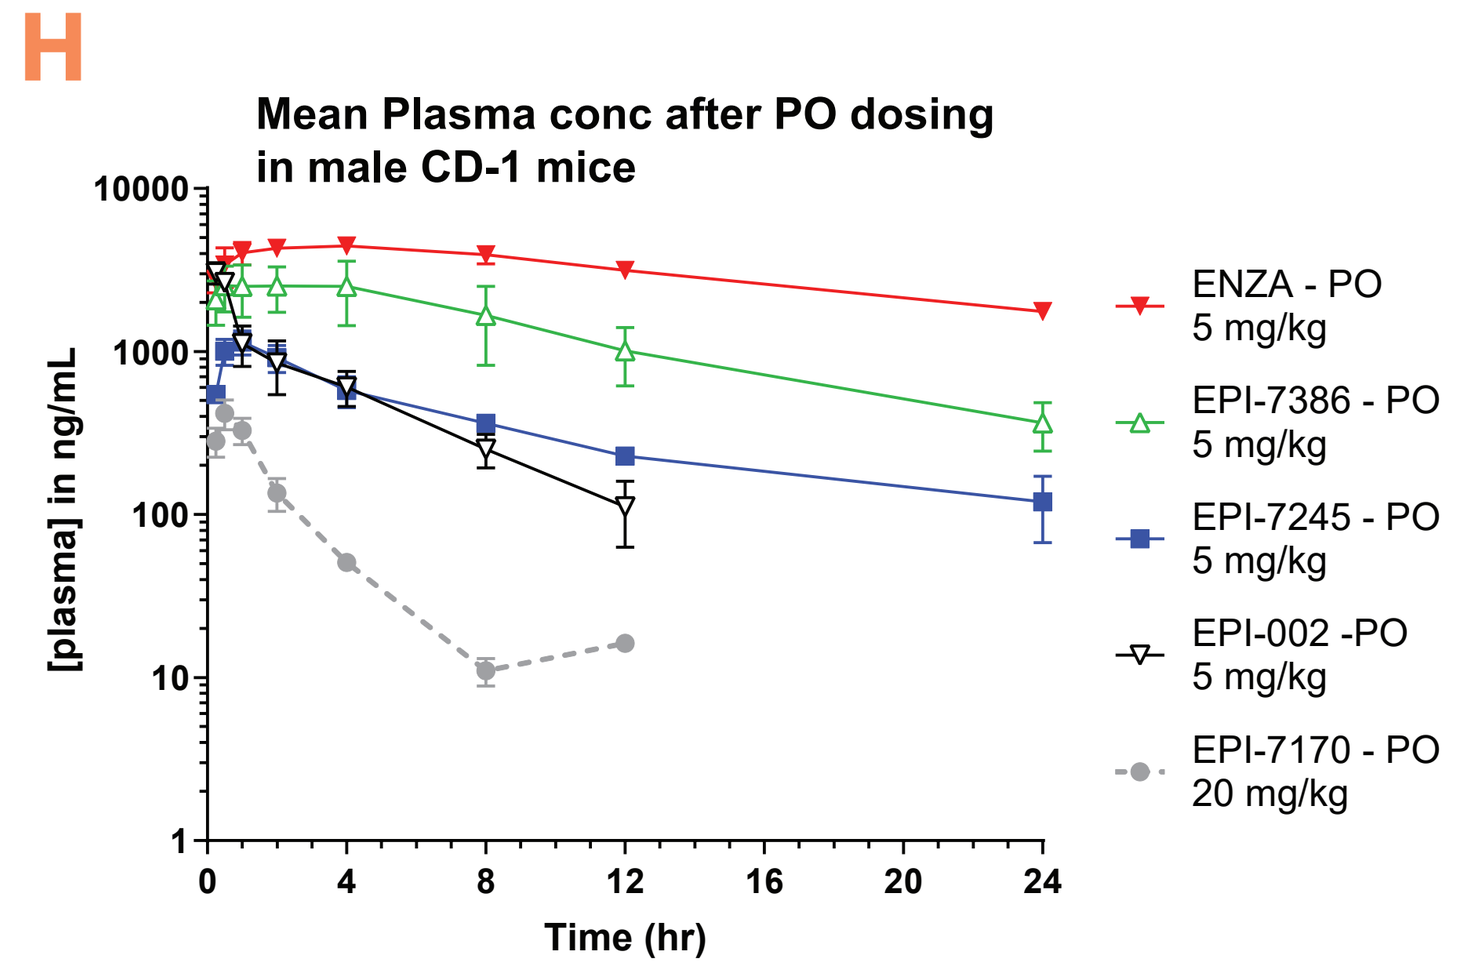

Mean Plasma conc after PO dosing in male CD-1 mice
[plasma] in ng/mL
Time (hr)
ENZA - PO 5 mg/kg
EPI-7386 - PO 5 mg/kg
EPI-7245 - PO 5 mg/kg
EPI-002 -PO 5 mg/kg
EPI-7170 - PO 20 mg/kg

I

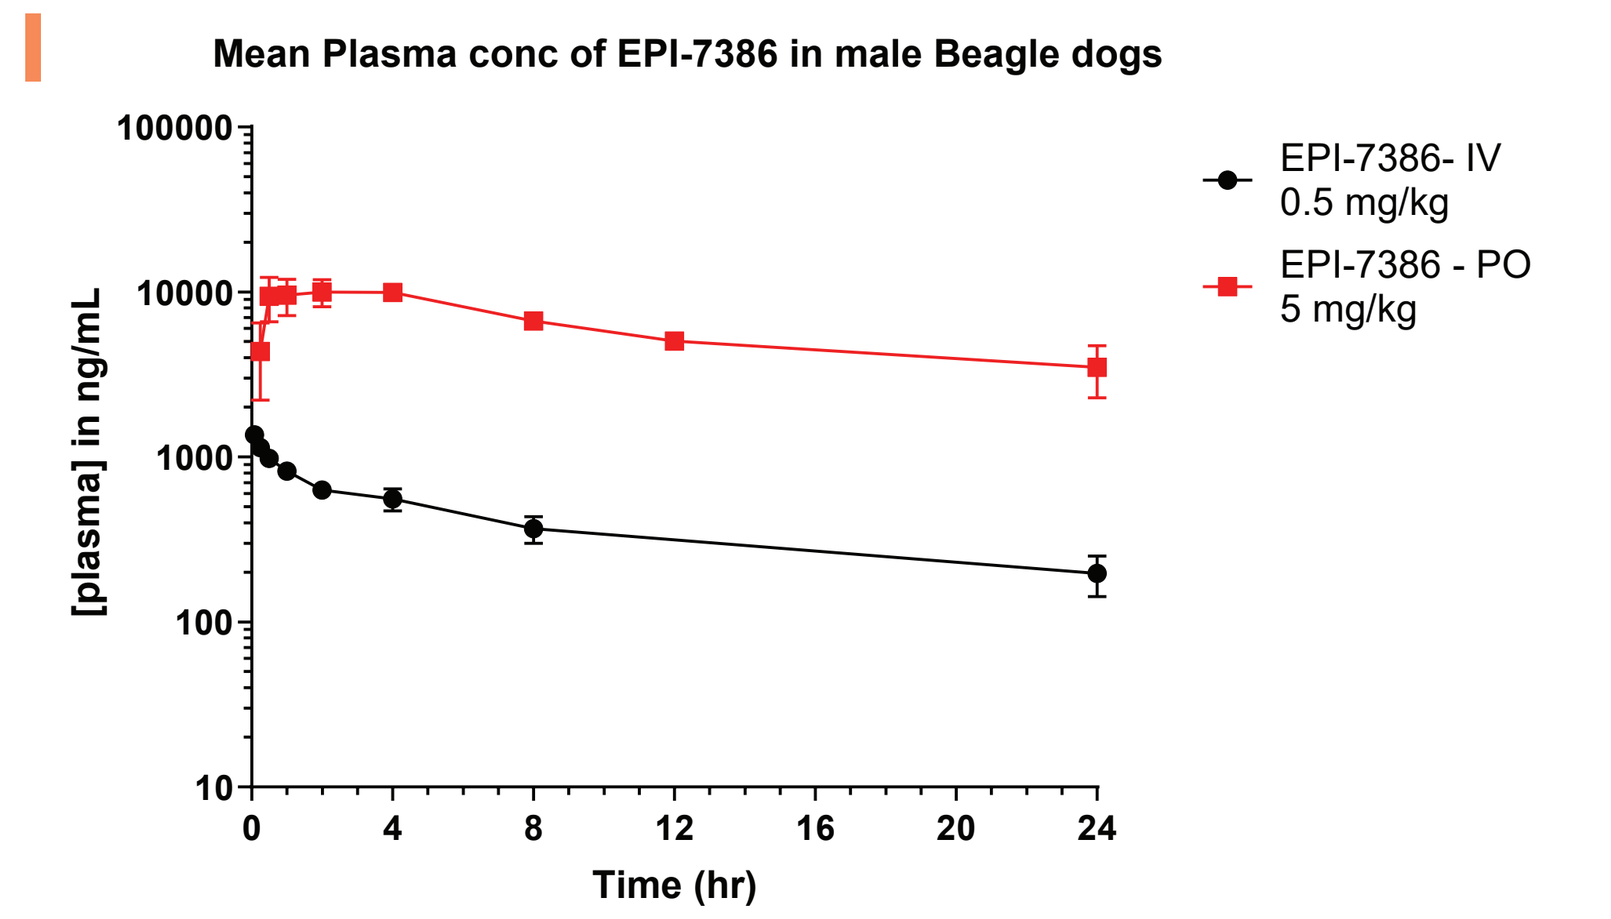

Mean Plasma conc of EPI-7386 in male Beagle dogs
[plasma] in ng/mL
Time (hr)
EPI-7386- IV 0.5 mg/kg
EPI-7386 - PO 5 mg/kg

Figure 4: Anitens are metabolically stable and show adequate PK profile for high and sustained plasma exposure
(A) Compound stability was assessed in human, mouse and rat hepatocytes. (B) Permeability of EPI-7386 was tested in Caco-2 assay at 1 uM. (C-D) Summary of IV and PO PK parameters after a single dose in male CD-1 mice (n=3). (E-F) Summary of IV and PO PK parameters after a single dose in male Beagle dogs. (G-H) PK curve after a single IV or PO dose in male CD-1 mice (n=3). EPI-7170 PO PK at 20 mg/kg was added as comparator. (I) PK curve after a single IV dose in male Beagle dogs (n=3).

Next generation Anitens are active and well tolerated in castrated mice bearing AR dependent xenografts

A

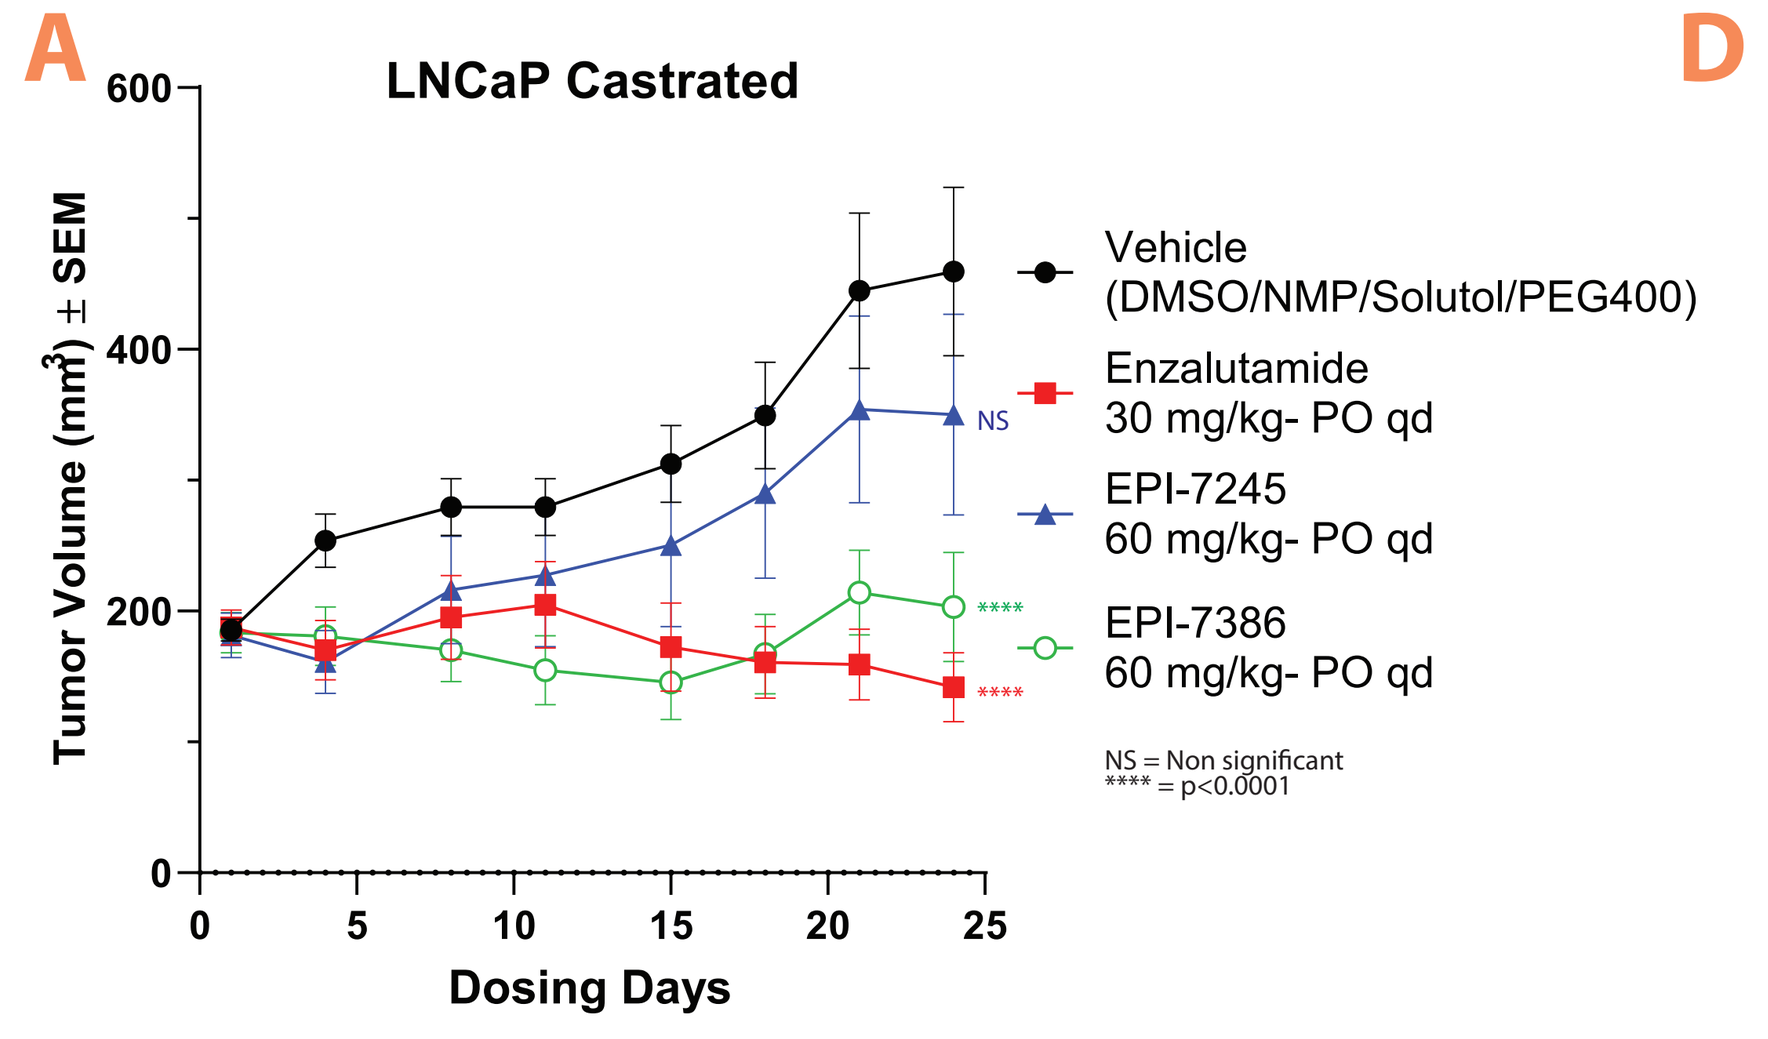

LNCaP Castrated
Tumor Volume (mm³) ± SEM
Dosing Days
Vehicle (DMSO/NMP/Solutol/PEG400)
Enzalutamide 30 mg/kg- PO qd
EPI-7245 60 mg/kg- PO qd
EPI-7386 60 mg/kg- PO qd
NS = Non significant
**** = p<0.0001

D

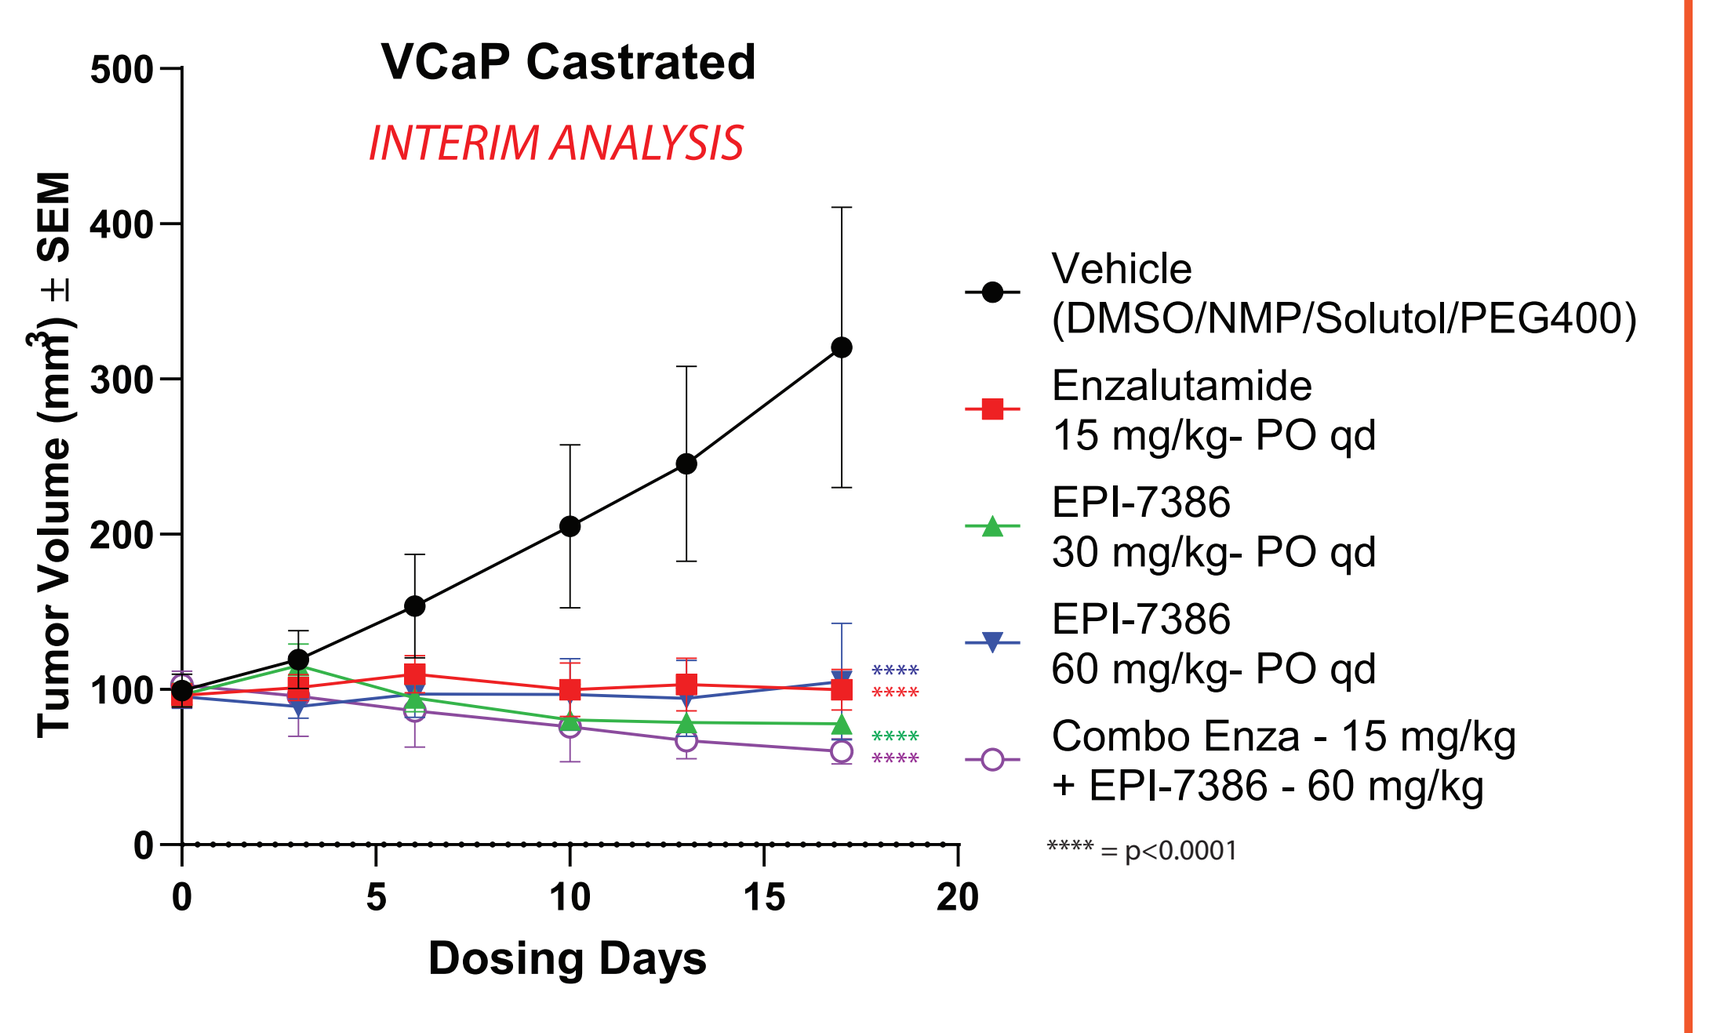

VCaP Castrated
INTERIM ANALYSIS
Tumor Volume (mm³) ± SEM
Dosing Days
Vehicle (DMSO/NMP/Solutol/PEG400)
Enzalutamide 15 mg/kg- PO qd
EPI-7386 30 mg/kg- PO qd
EPI-7386 60 mg/kg- PO qd
Combo Enza - 15 mg/kg + EPI-7386 - 60 mg/kg
**** = p<0.0001

B

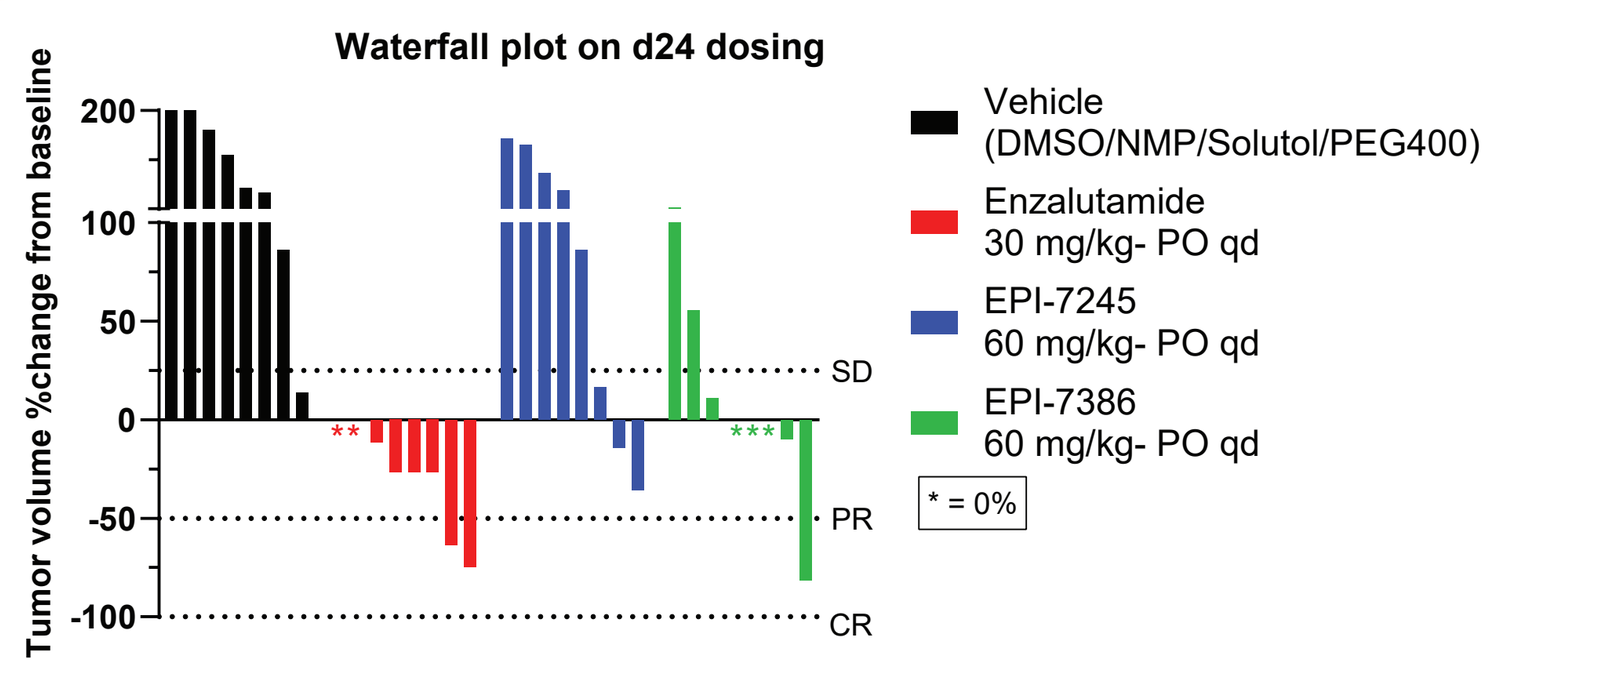

Waterfall plot on d24 dosing
Tumor volume %change from baseline
SD
PR
CR
Vehicle (DMSO/NMP/Solutol/PEG400)
Enzalutamide 30 mg/kg- PO qd
EPI-7245 60 mg/kg- PO qd
EPI-7386 60 mg/kg- PO qd
* = 0%

E

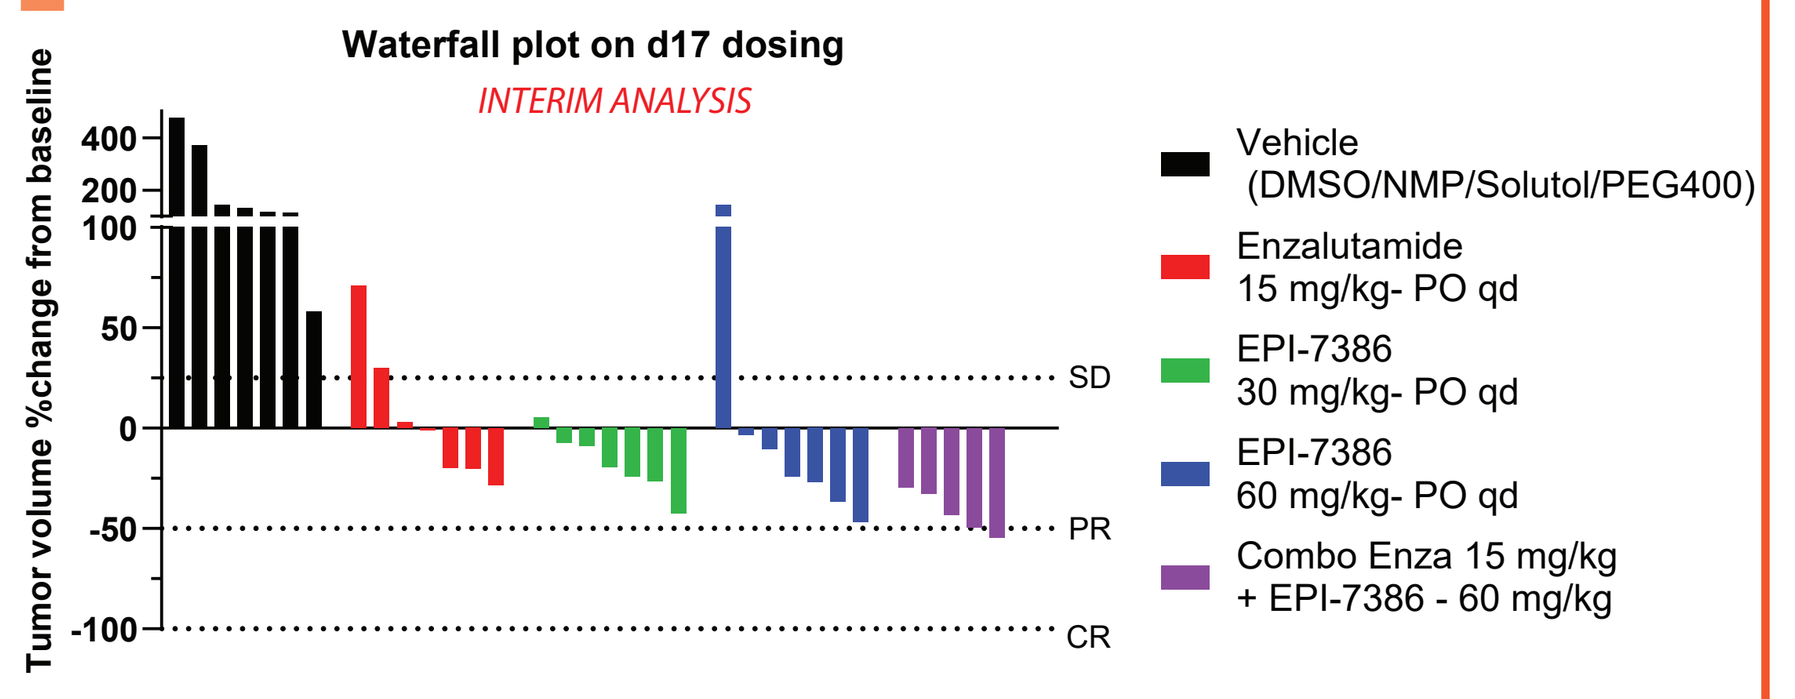

Waterfall plot on d17 dosing
INTERIM ANALYSIS
Tumor volume %change from baseline
SD
PR
CR
Vehicle (DMSO/NMP/Solutol/PEG400)
Enzalutamide 15 mg/kg- PO qd
EPI-7386 30 mg/kg- PO qd
EPI-7386 60 mg/kg- PO qd
Combo Enza 15 mg/kg + EPI-7386 - 60 mg/kg

C

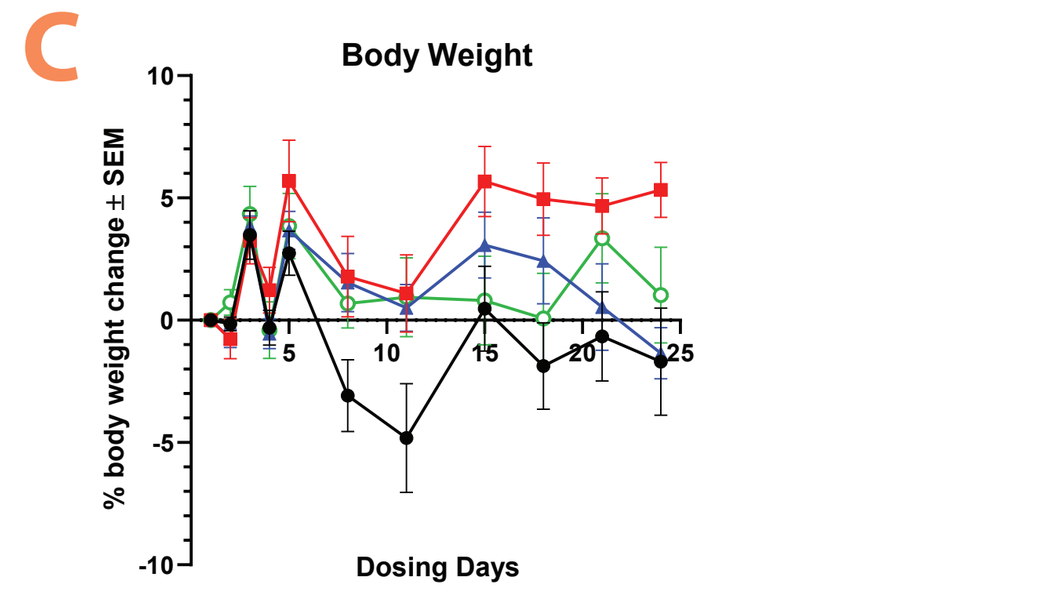

Body Weight
% body weight change ± SEM
Dosing Days

F

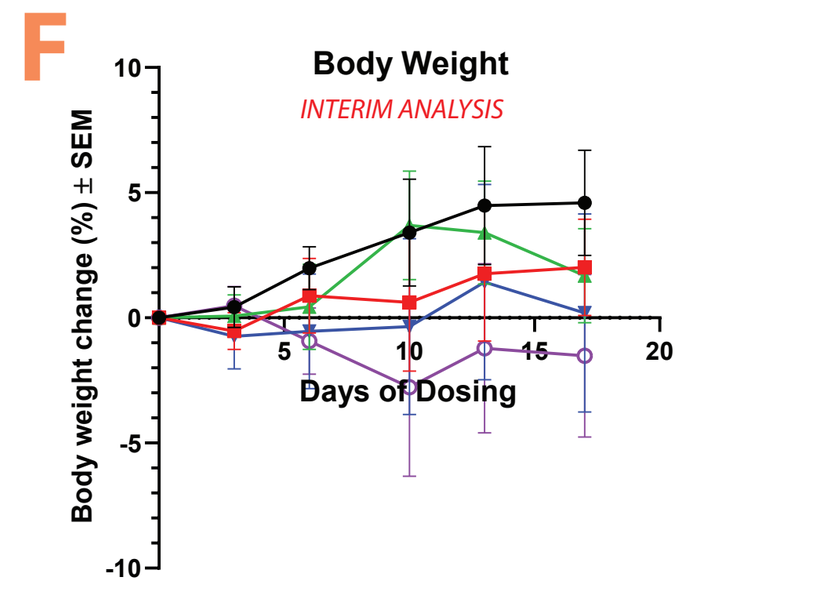

Body Weight
INTERIM ANALYSIS
Body weight change (%) ± SEM
Days of Dosing

G

Compound name	Route	Dose (mg/kg)	Cmax (ng/mL)	Tmax (h)	T1/2 (h)	AUC0-last (ng·h/mL)
EPI-7386	PO	60.00	41,667	4	5.4	482,822
Enzalutamide	PO	15.0	21,067	9	ND	388,202
EPI-7386 combo	PO	60.00	51,900	3	6.9	603,960
Enzalutamide combo	PO	15.0	11,467	12	ND	231,974

Figure 5: In vivo activity in CRPC xenograft models
(A) Tumor growth in male NCG mice bearing LNCaP tumors. Castration was performed when tumors reached ~ 100 mm³ and dosing started 1 week after. (B) Individual tumor volume change measured on day 24. (C) The body weight change showed no drug-related toxicity. (D) Tumor growth measured in male SCID Beige mice bearing VCaP tumors. Castration was performed when tumors reached ~ 100 mm³ and dosing started 2 weeks after. (E) Individual tumor volume change measured on day 17. (F) The body weight was captured biweekly and normalized to baseline. (G) PK parameters of EPI-7386 or enzalutamide dosed as single agent or combination in CD-1 male mice.

CONCLUSION

- Promising next-generation Aniten compounds have been identified and display:
 a. Similar potency in vitro to the 'lutamides
 b. Activity in several in vitro cell lines, including enzalutamide resistant models
 c. On-target NTD inhibition with minimal activity against other nuclear hormone receptors
 d. No PXR agonist activity and no inhibition of the GABA-chloride channel which is responsible for inducing seizures in enzalutamide or apalutamide treated patients
 e. Favorable metabolic profile in vitro (Caco-2 and hepatocytes)
 f. Favorable in vivo metabolic profile with high exposure and long half-life
 g. Comparable activity to enzalutamide in AR dependent LNCaP and VCaP xeno grafts
 h. Qualitatively improved response in combination with enzalutamide
- **Based upon the above criteria, EPI-7386 selected as the IND candidate with phase 1 study anticipated 1Q 2020**